First-in-human Phase I Study of the Bifunctional EGFR/TGFβ Fusion Protein BCA101 in Patients with EGFR-driven Advanced Solid Cancers

Filip Janku, Glenn J. Hanna, Richard D. Carvajal, Paul K. Paik, Alberto Hernando-Calvo, Maura L. Gillison, Siqing Fu, Jennifer J. Wheler, David Bohr, Ralf Reiners, Seng-Lai Tan, Sanela Bilic, Philippe L. Bedard; The University of Texas MD Anderson Cancer Center, Houston, TX; Dana-Farber Cancer Institute, Boston, MA; Columbia University Irving Medical Center, New York, NY; Memorial Sloan-Kettering Cancer Center, New York, NY; Princess Margaret Cancer Centre, University of Toronto, Toronto, ON; Bicara Therapeutics, Cambridge, MA; Vanadro, LLC, Urbandale, IA

Background and Rationale

Figure 1: BCA101, a bispecific antibody targeting EGFR and TGFβ

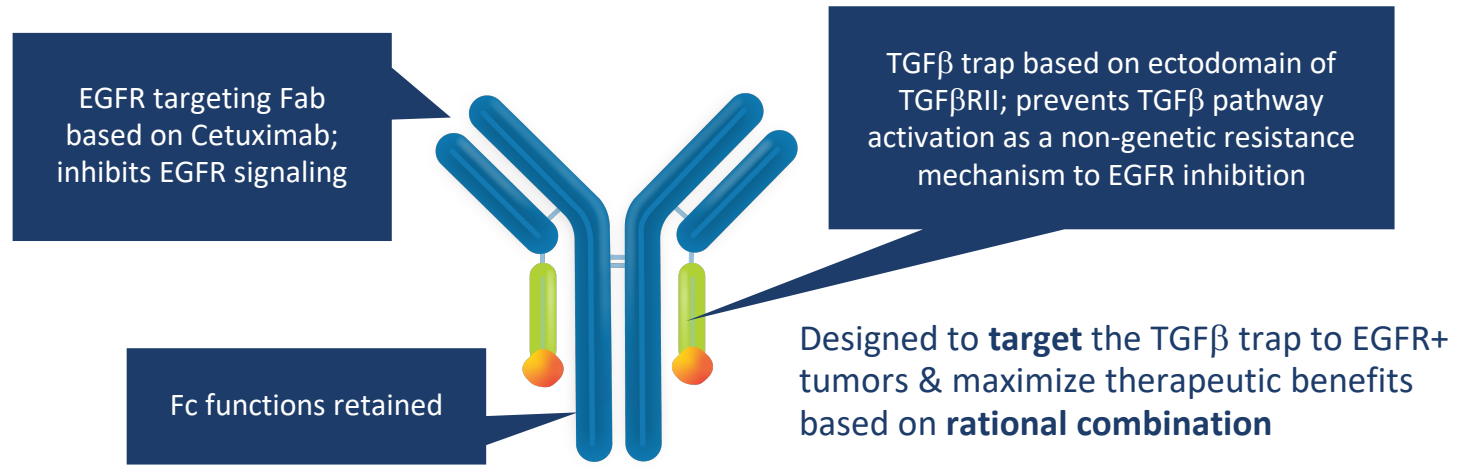
EGFR targeting Fab based on Cetuximab; inhibits EGFR signaling
TGFβ trap based on ectodomain of TGFβRII; prevents TGFβ pathway activation as a non-genetic resistance mechanism to EGFR inhibition
Fc functions retained
Designed to **target** the TGFβ trap to EGFR+ tumors & maximize therapeutic benefits based on **rational combination**

Figure 2: Combined mechanisms of action of BCA101

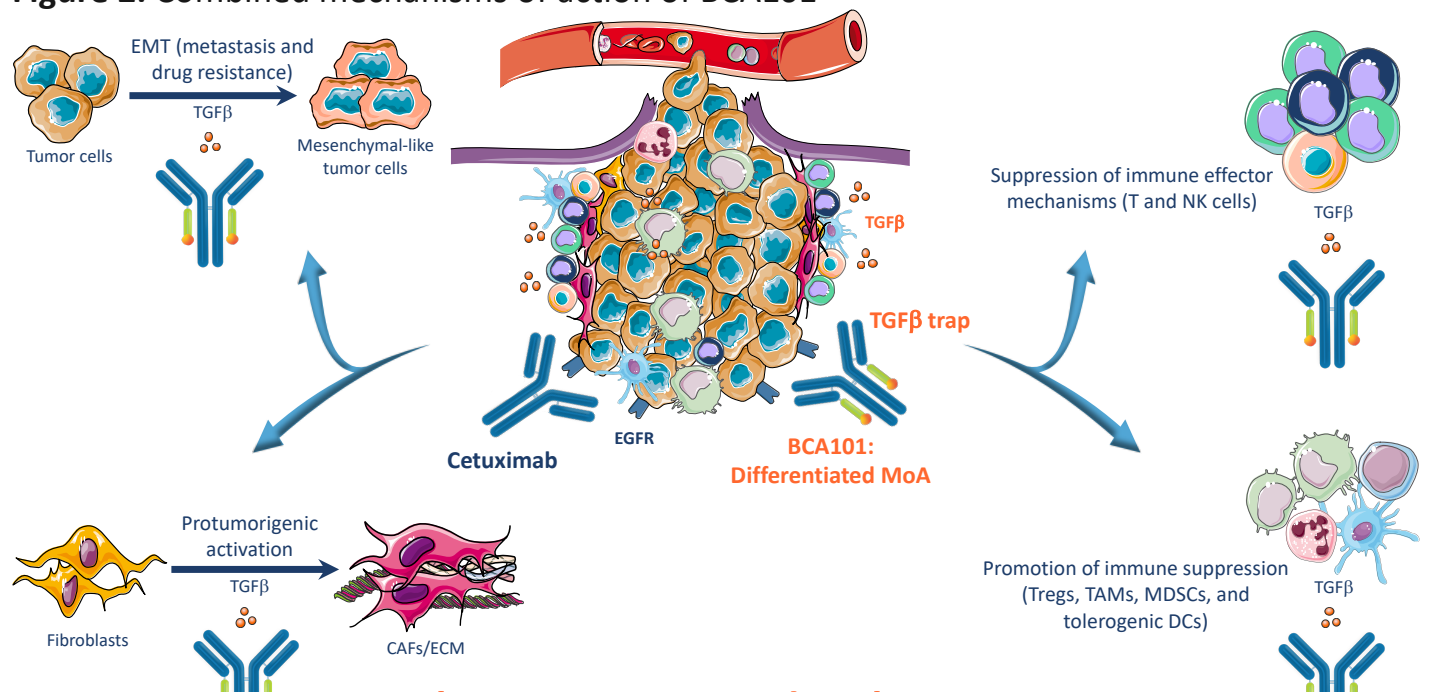
Tumor cells
EMT (metastasis and drug resistance)
TGFβ
Mesenchymal-like tumor cells
TGFβ
TGFβ trap
Cetuximab
EGFR
BCA101: Differentiated MoA
Suppression of immune effector mechanisms (T and NK cells)
TGFβ
Fibroblasts
Protumorigenic activation
TGFβ
CAFs/ECM
Promotion of immune suppression (Tregs, TAMs, MDSCs, and tolerogenic DCs)
TGFβ

Trial Design and Objectives

Key Inclusion Criteria

- Histologically or cytologically confirmed, EGFR-driven, advanced solid tumors refractory to current standard of care therapy
- Must have evaluable or measurable disease per RECIST v1.1 and iRECIST

Primary Objective:

- Characterize safety and tolerability of SA BCA101 and BCA101 in combination with pembrolizumab and to establish MTD and/or RD

Secondary Objectives:

- Characterize PK profile and evaluate immunogenicity in single agent BCA101 and combination with pembrolizumab
- Evaluate preliminary anti-tumor effects

Exploratory Objectives:

- Explore pharmacodynamic and biomarkers

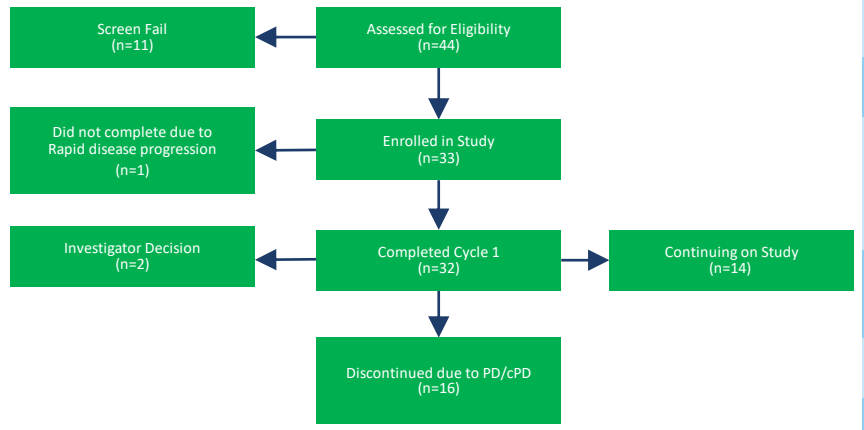
Screen Fail (n=11)
Assessed for Eligibility (n=44)
Did not complete due to Rapid disease progression (n=1)
Enrolled in Study (n=33)
Investigator Decision (n=2)
Completed Cycle 1 (n=32)
Continuing on Study (n=14)
Discontinued due to PD/cPD (n=16)

Subject Demography and Exposure (N=33)

Age (y)	Mean (Range)	61.2 (47-77)
Sex (n)	Male / Female	18/15
Race (n)	White	28
	Asian	2
	Not reported	3
Prior lines of systemic therapy (n)	Mean (Range)	4 (1-8)
Performance status	ECOG 0/1	9/24
Number of BCA101 doses	Mean (Range)	6.1 (1-18)

Data as of 1 April 2021

BCA101X1101 (NCT04429542) is a phase 1/1b, open-label, multicenter study to evaluate the safety, tolerability, pharmacokinetics, pharmacodynamics and anti-tumor activity of BCA101

Figure 3: BCA101 is well tolerated with no DLTs observed

BCA101X1101: Subjects with Treatment Related AEs (TRAEs)

Adverse Events in > 1 Subject	Maximum Grade per Patient				
31 of 33 Subjects with ≥1 related events	1	2	3	4	Total (%)
Dermatitis acneiform	12	6			18 (54.5)
Fatigue	7				7 (21.2)
Nausea	4				4 (12.1)
Chills	2	1			3 (9.1)
Fever	2	1			3 (9.1)
Hypophosphatemia		2	1		3 (9.1)
Lipase Increase	2	1			3 (9.1)
Rash	2	1			3 (9.1)
Stomatitis	3				3 (9.1)
Aspartate aminotransferase increased	2				2 (6.1)
Diarrhea	2				2 (6.1)
Epistaxis	2				2 (6.1)
Headache	2				2 (6.1)
Pruritis	2				2 (6.1)
Rash maculo-papular	1	1			2 (6.1)

Data as of 1 April 2021

Figure 4: BCA101 achieves dose-proportional exposure

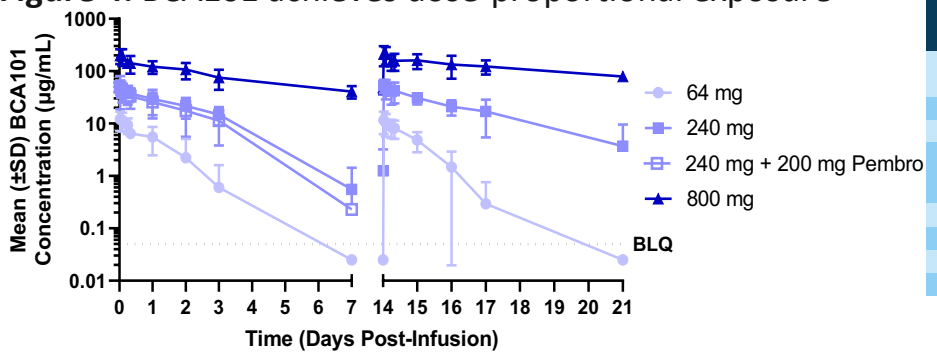
Mean (±SD) BCA101 Concentration (µg/mL)
1000
100
10
1
0.1
0.01
BLQ
0 1 2 3 4 5 6 7 14 15 16 17 18 19 20 21
Time (Days Post-Infusion)
64 mg
240 mg
240 mg + 200 mg Pembro
800 mg

PK Parameters

Dose (mg)	AUC_{inf} (hr*µg/mL)	C_{max} (µg/mL)	$T_{1/2}$ (hr)	CL (L/hr)	V_z (L)
64	340	12.1	14	0.242	4.2
240	2796	52.8	31	0.091	4.1
240 (+ 200 mg Pembro)	2185	50.5	27	0.131	5.5
500*	8922	136	49	0.073	4.9
750*	10831	156	62	0.075	6.4
800	19260	216	91	0.045	5.9
1000*	15440	231	68	0.069	6.6

*Not plotted due to insufficient data from ongoing cohorts

Figure 5: BCA101 preliminary anti-tumor activity

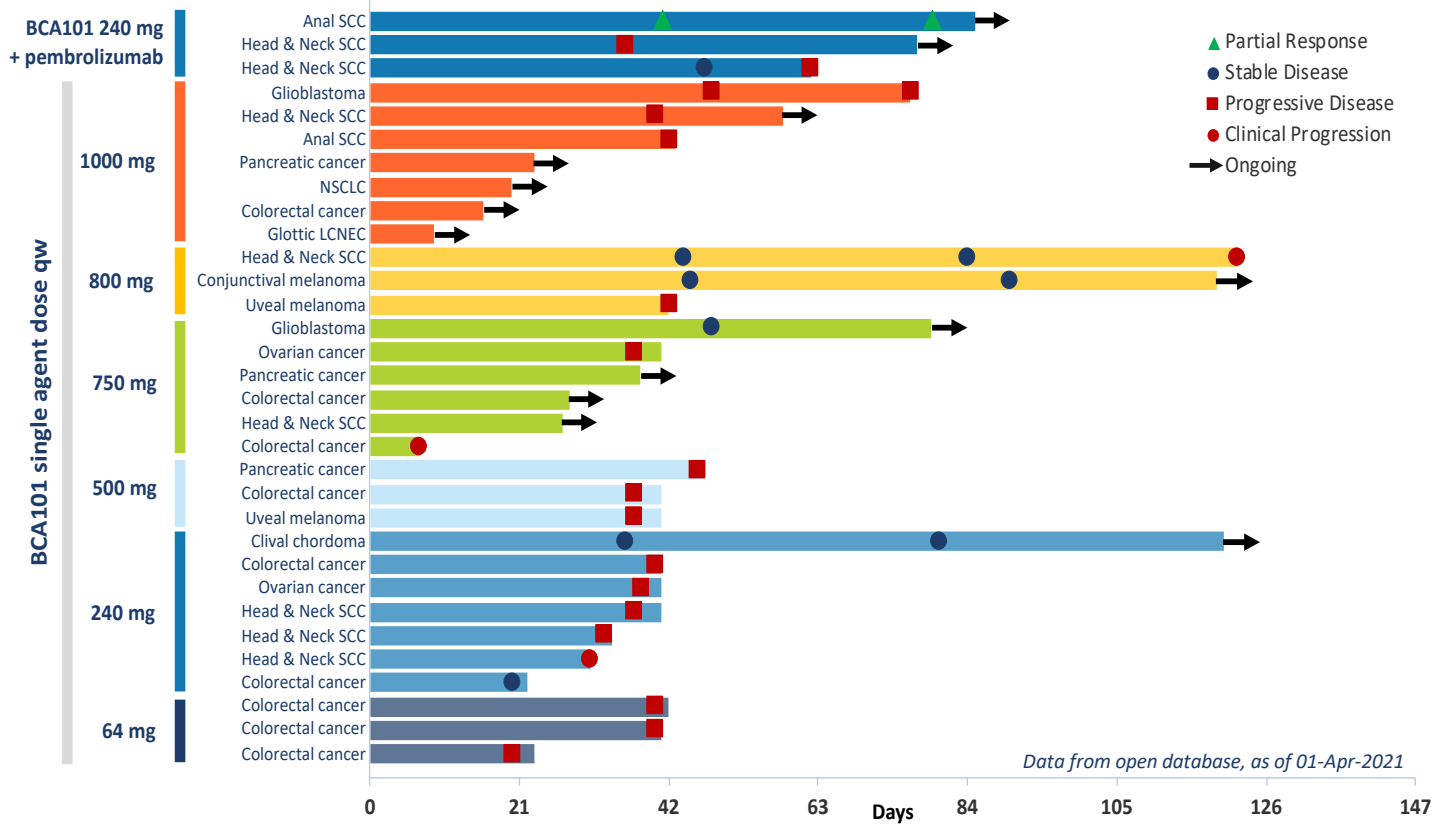
BCA101 240 mg + pembrolizumab
BCA101 single agent dose qw
1000 mg
800 mg
750 mg
500 mg
240 mg
64 mg
Anal SCC
Head & Neck SCC
Head & Neck SCC
Glioblastoma
Head & Neck SCC
Anal SCC
Pancreatic cancer
NSCLC
Colorectal cancer
Glottic LCNEC
Head & Neck SCC
Conjunctival melanoma
Uveal melanoma
Glioblastoma
Ovarian cancer
Pancreatic cancer
Colorectal cancer
Head & Neck SCC
Colorectal cancer
Pancreatic cancer
Colorectal cancer
Uveal melanoma
Clival chordoma
Colorectal cancer
Ovarian cancer
Head & Neck SCC
Head & Neck SCC
Head & Neck SCC
Colorectal cancer
Colorectal cancer
Colorectal cancer
Colorectal cancer
Partial Response
Stable Disease
Progressive Disease
Clinical Progression
Ongoing
Data from open database, as of 01-Apr-2021
0 21 42 63 84 105 126 147
Days

Figure 6: BCA101 completely sequesters TGFβ1 and partially neutralizes TGFβ2 at ≥500mg in plasma

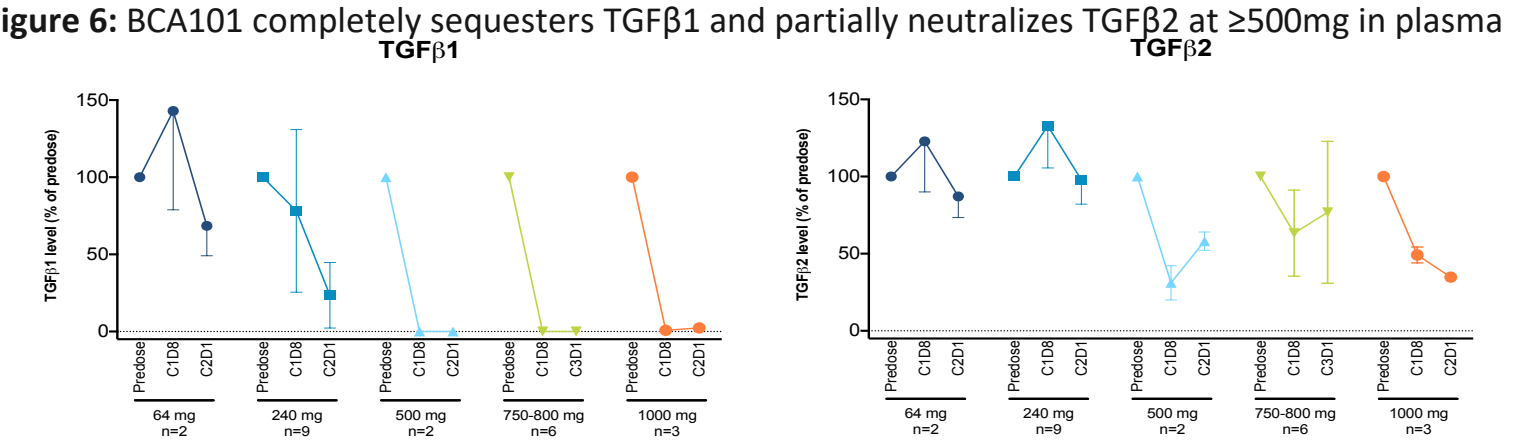
TGFβ1
TGFβ2
TGFβ1 level (% of predose)
TGFβ2 level (% of predose)
Predose C1D8 C2D1
64 mg n=2
240 mg n=9
500 mg n=2
750-800 mg n=6
1000 mg n=3

Figure 7: BCA101 increases T cell populations and decreases granulocytic MDSCs in the peripheral blood

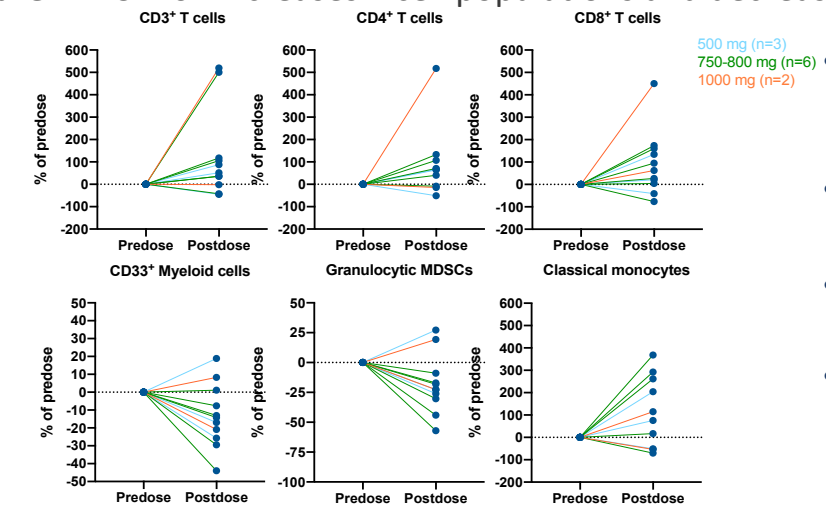
CD3+ T cells
CD4+ T cells
CD8+ T cells
CD33+ Myeloid cells
Granulocytic MDSCs
Classical monocytes
% of predose
Predose Postdose
500 mg (n=3)
750-800 mg (n=6)
1000 mg (n=2)

- We assessed changes in circulating immune cell profiles pre- and post-BCA101 treatment by flow cytometry.
- BCA101 significantly increased the percentages of T cells and classical monocytes.
- Conversely, BCA101 significantly decreased the percentages of granulocytic MDSCs.
- Blood immune phenotyping data suggest immunopotentiating responses to BCA101 treatment.

Figure 8: BCA101 demonstrates target engagement at the tumor site: BCA101 decreases SMAD2-phosphorylation in tumor tissues

- Phospho-SMAD2 is a main downstream effector of the TGFβ pathway and, therefore, indicates the activation of this signaling axis in tumors
- We assessed the changes in SMAD2 activation in tumor tissues pre- and post-treatment with BCA101 using immunohistochemistry.
- Preliminary data suggest that treatment with BCA101 at higher doses reduces phospho-SMAD2 signal in tumors.

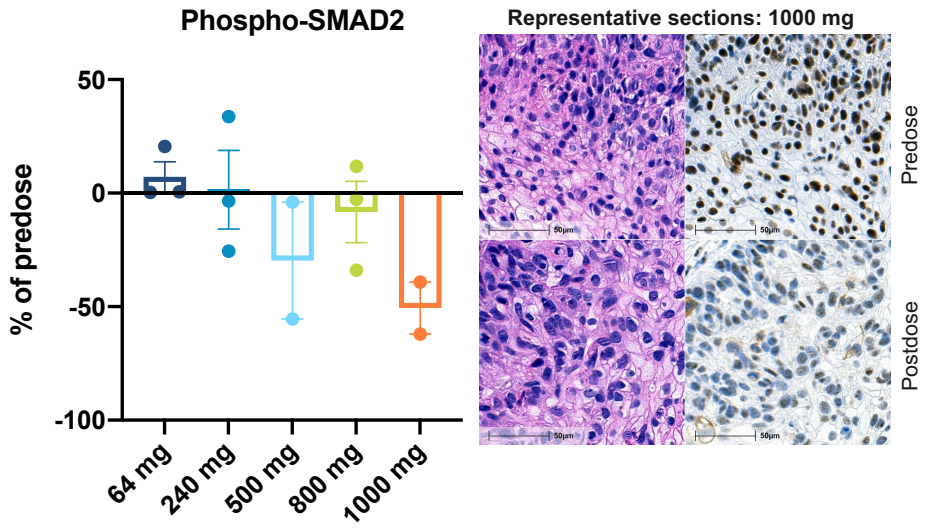
Phospho-SMAD2
% of predose
50
0
-50
-100
64 mg
240 mg
500 mg
800 mg
1000 mg
Representative sections: 1000 mg
Predose
Postdose

Conclusions

- The highest tested dose level as of 1 April 2021 is 1000mg in single agent and continues to be well tolerated. Rash, is the most frequent TRAE. The maximum tolerated dose has not been reached.
- One partial response has been observed in combination with pembrolizumab in a PD-(L)1 naïve subject with cancer of the anal canal.
- BCA101 achieves dose-proportional PK and demonstrates target engagement based on pharmacodynamic biomarkers in both plasma and tissue
- BCA101 dose escalation continues, currently at the 1250mg dose level in single agent and 750mg dose level in combination with pembrolizumab.

Acknowledgements: The authors would like to thank Liviu Niculescu and Amir Al-Khami from Bicara Therapeutics for their contributions.