Development of BCA101, a Bifunctional Antibody Capable of Simultaneously Disabling EGFR and TGF-β Signaling, as a Novel Single-agent Immunotherapy

Srinivas Reddy Boreddy[1], Reshmi Nair[1], Arindam Banerjee[1], Anshu Kuriakose[1], Prashant Kumar Pandey[1], Chaitali Dey[1], Meena Shri[1], Shruthi Rao[1], Shiva Kuman B M[1], Moni Abraham Kuriakose[2], Ram Bhupal Reddy[2], Amrita Suresh[2], Praveen Reddy Moole[1], Usha Bhugani[1], Seng-Lai Tan[3], Pradip Nair[1]

Author Affiliations: [1] –Biofusion Therapeutics, Bengaluru, India; [2] –Mazumdar-Shaw Medical Centre, Bengaluru, India; [3] –Bicara Therapeutics, Cambridge, USA

Background

Given the pleiotropic functions of transforming growth factor-beta (TGF-β), current approaches to targeting systemic TGF-β will likely lead to suboptimal clinical activity and/or undesirable effects such as acute bleeding and cardiotoxicity. Epidermal growth factor receptor (EGFR) is one of the most extensively validated tumor-associated antigens. Bicara Therapeutics has developed a novel bifunctional fusion protein, composed of a monoclonal antibody against EGFR and an extracellular domain of human TGF-β receptor II (TGF-βRII). We demonstrate BCA101 has the potential to improve anti-tumor response by leveraging the cooperativity between EGFR and TGF-β signaling pathways by predominantly neutralizing TGFβ in EGFR-expressing tumors.

Figure 1: Combined mechanisms of action of BCA101

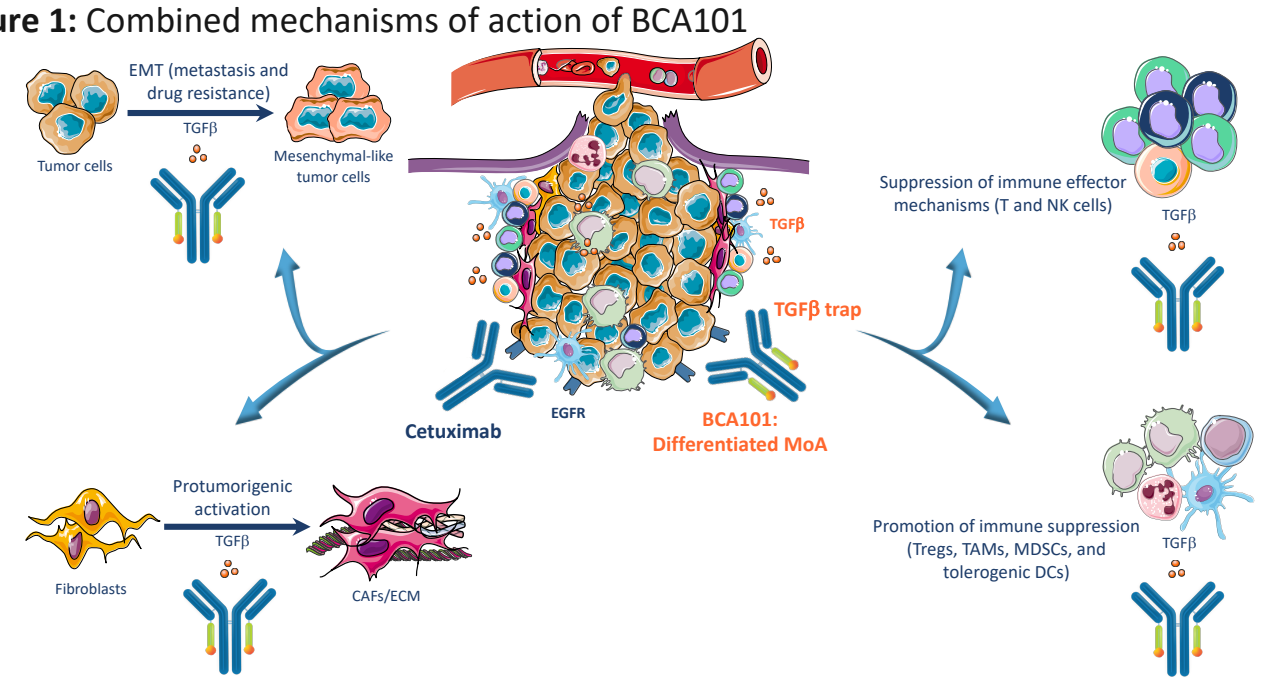
Tumor cells
EMT (metastasis and drug resistance)
TGFβ
Mesenchymal-like tumor cells
TGFβ
TGFβ trap
Cetuximab
EGFR
BCA101: Differentiated MoA
Suppression of immune effector mechanisms (T and NK cells)
TGFβ
Fibroblasts
Protumorigenic activation
TGFβ
CAFs/ECM
Promotion of immune suppression (Tregs, TAMs, MDSCs, and tolerogenic DCs)
TGFβ

Figure 2: BCA101, a bifunctional antibody targeting EGFR and TGF-β

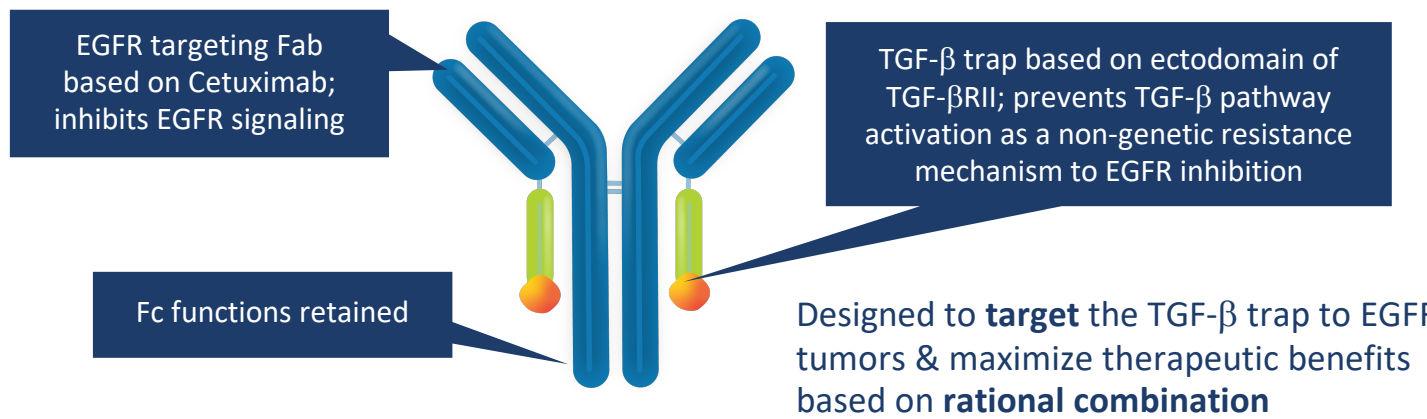
EGFR targeting Fab based on Cetuximab; inhibits EGFR signaling
TGF-β trap based on ectodomain of TGF-βRII; prevents TGF-β pathway activation as a non-genetic resistance mechanism to EGFR inhibition
Fc functions retained

Designed to **target** the TGF-β trap to EGFR+ tumors & maximize therapeutic benefits based on **rational combination**

Figure 3: BCA101 is a bifunctional protein designed to target both EGFR and TGF-β simultaneously

Sample	KD (nM)	SD
EGFR Binding (SPR)		
BCA101	2.58	0.63
Cetuximab	1.16	0.19
TGF-β1 Binding (SPR)		
BCA101	61.3	13.29
TGF-βRIIFc	64.8	7.35

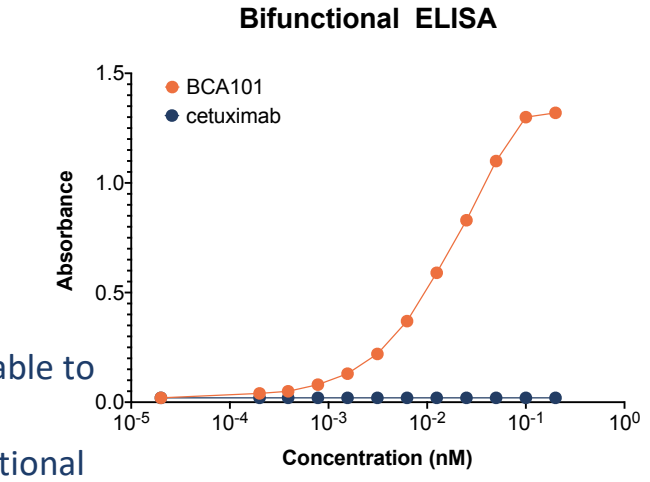
Bifunctional ELISA
BCA101
cetuximab
Absorbance
Concentration (nM)

- EGFR and TGF-β target binding affinities were comparable to cetuximab and TGF-βRIIECD, respectively
- Both the arms are simultaneously functional in bi-functional ELISA

Figure 4: Improved retention of BCA101 in tumor microenvironment (TME)

- Relatively, BCA101 stays longer in TME compared to TGF-βRII-Fc
- BCA101 neutralizes 50% mouse serum TGF-β1 at 0.2 μg/mL (EC50)
- Tumor PK analysis showed that BCA101 at a dose of 10 mg/kg suppressed tumor TGFβ1 levels by about 90%, whereas TGF-βRII-Fc at equimolar dose showed about 54% suppression (data not shown)

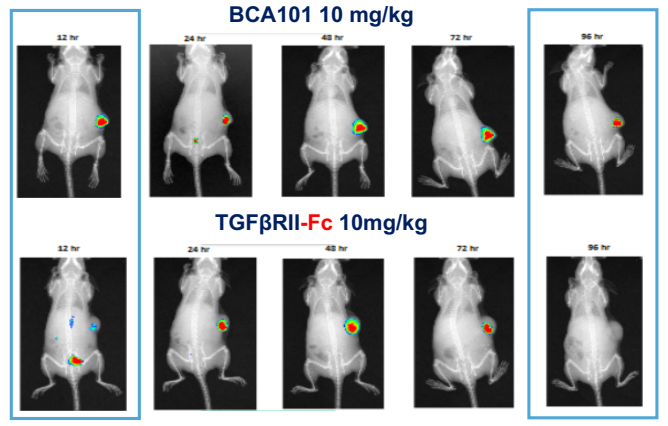
BCA101 10 mg/kg
TGFβRII-Fc 10mg/kg

Figure 5: EGFR and TGF-β targeting arms and Fc of BCA101 are functionally active

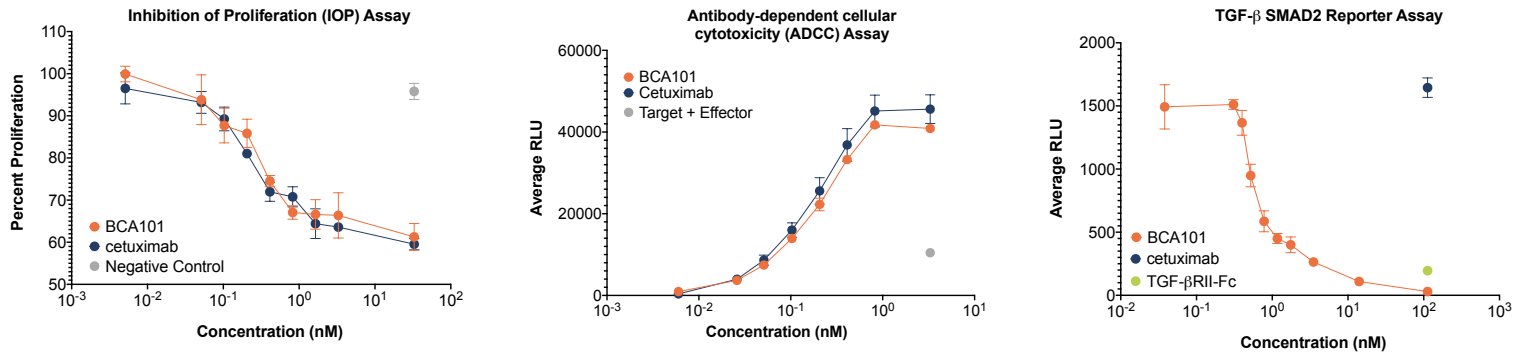
Inhibition of Proliferation (IOP) Assay
Percent Proliferation
BCA101
cetuximab
Negative Control
Concentration (nM)
Antibody-dependent cellular cytotoxicity (ADCC) Assay
Average RLU
BCA101
Cetuximab
Target + Effector
Concentration (nM)
TGF-β SMAD2 Reporter Assay
Average RLU
BCA101
cetuximab
TGF-βRII-Fc
Concentration (nM)

- BCA101 anti-EGFR activity was measured in inhibition of proliferation (IOP) assay; antibody-dependent cellular cytotoxicity (ADCC) & TGF-β activity was measured by reporter assays in multiple cell lines
- BCA101 IOP and ADCC activity was comparable to cetuximab
- BCA101 neutralizes TGF-β1 and TGF-β3 but not TGF-β2 in TGF-β SMAD reporter assay, showing comparable activity to TGF-βRII-Fc

Cell line	BCA101	Cetuximab
IOP EC_{50} Mean ± SD (nM)		
FaDu	0.08 ± 0.02	0.06 ± 0.03
NCI H508	0.15 ± 0.07	0.09 ± 0.03
CAL27	0.33 ± 0.20	0.23 ± 0.06

Cell line	BCA101	Cetuximab
ADCC EC_{50} Mean ± SD (nM)		
FaDu	0.14 ± 0.02	0.11 ± 0.00
CAL27	0.23 ± 0.05	0.17 ± 0.03
A431	0.22 ± 0.12	0.15 ±0.05

Figure 6: BCA101 inhibits TGF-β-induced epithelial to mesenchymal transition (EMT)

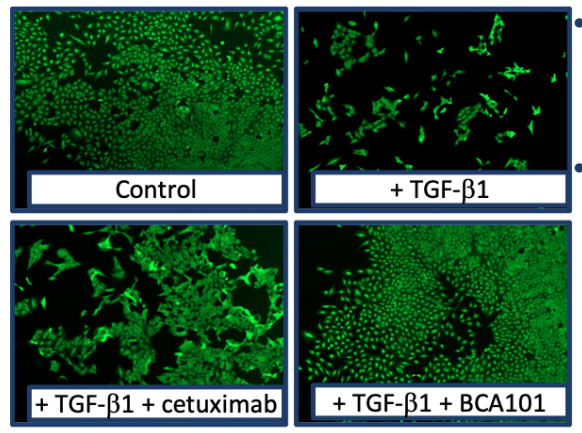
Control
+ TGF-β1
+ TGF-β1 + cetuximab
+ TGF-β1 + BCA101

- BCA101, but not cetuximab, rescued TGF-β1-induced cell invasion and EMT markers (Vimentin & E-cadherin) in 2D cultures and 3D spheroid assays
- BCA101 rescued TGF-β1 plus EGF induced VEGF secretion in Spheroids

Figure 7: BCA101 rescues from TGF-β-mediated immune inhibition

- TGFβ-1 suppressed NKG2D expression on CD8+ T-cells and increased percentage of CD4+CD25+FoxP3+ cells (iTregs) in stimulated PBMCs
- BCA101, but not cetuximab, significantly rescued NKG2D expression on CD8+ T cells and decreased iTreg population, indicating the immune activation

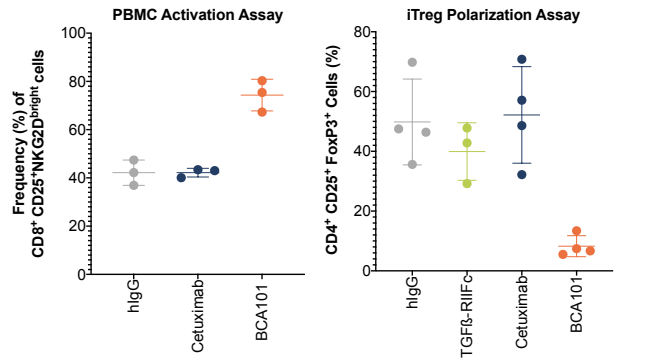
PBMC Activation Assay
hIgG
Cetuximab
BCA101
iTreg Polarization Assay
hIgG
TGFβ-RIIFc
Cetuximab
BCA101

Figure 8: BCA101 demonstrates superior anti-tumor efficacy compared to cetuximab + TGF-βRII-Fc *in vivo*

- BCA101 demonstrated statistically significant improvement in anti-tumor efficacy compared to the combination of cetuximab + TGF-βRII-Fc in HNSCC Fadu xenograft study
- Study illustrates the synergy of the BCA101 molecule in localizing TGF-β neutralization in tumor microenvironment (TME) for improved efficacy

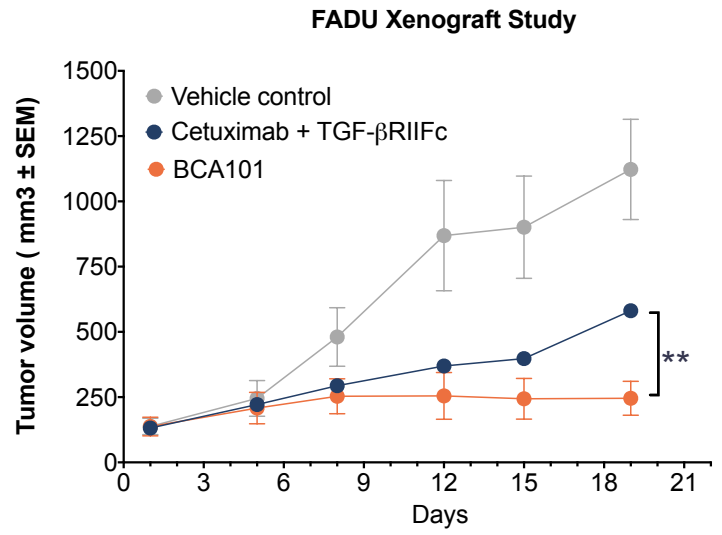
FADU Xenograft Study
Vehicle control
Cetuximab + TGF-βRIIFc
BCA101
Tumor volume (mm3 ± SEM)
Days
**

Figure 9: BCA101 demonstrates sustained tumor regression and delayed tumor relapse compared to cetuximab in HNSCC-PDX models

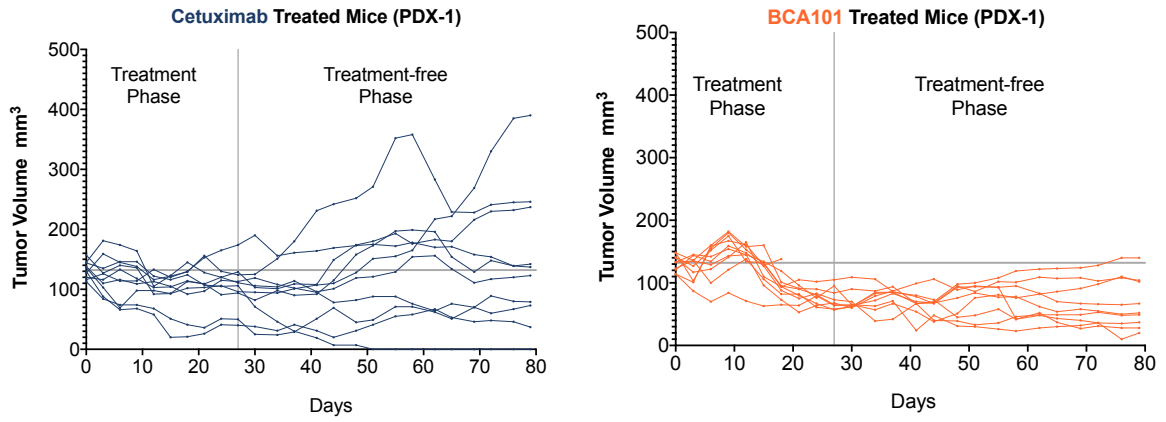
Cetuximab Treated Mice (PDX-1)
Treatment Phase
Treatment-free Phase
Tumor Volume mm3
Days
BCA101 Treated Mice (PDX-1)
Treatment Phase
Treatment-free Phase
Tumor Volume mm3
Days

- HNSCC PDX-bearing NSG mice were treated with BCA101 (10 mg/kg) or molar equivalent cetuximab for 27 days and left untreated until Day 79. During treatment-free / relapse phase, 6/10 tumors relapsed in cetuximab group and only 1/9 tumors in BCA101

(p = 0.04, Fisher Exact test)

Figure 10: BCA101 exhibited improved efficacy in combination with anti-PD1 inhibitor in checkpoint-resistant HuNOG-EXL humanized model

- PC-3 cell line chosen as known to be resistant to cetuximab & anti-PD1
- BCA101 + anti-PD1 demonstrated statistically significant anti-tumor efficacy compared to anti-PD1 or BCA101 alone
- Demonstrates rationale for combination therapy approach in checkpoint-inhibitor-resistant tumors

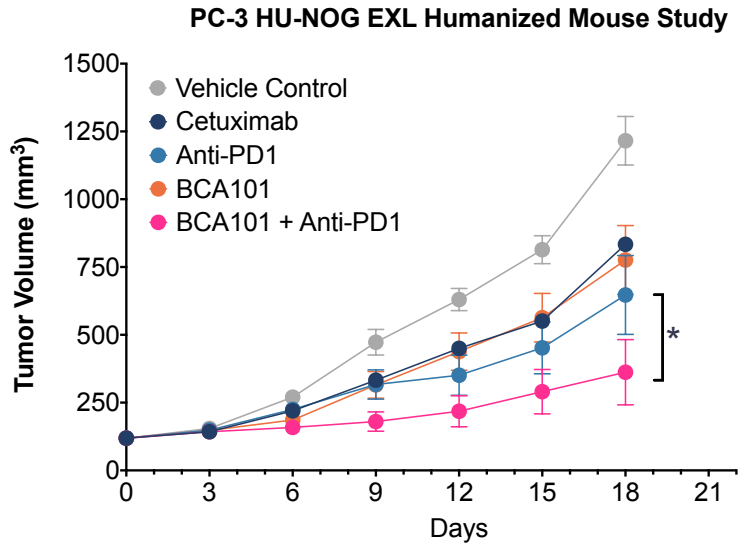
PC-3 HU-NOG EXL Humanized Mouse Study
Vehicle Control
Cetuximab
Anti-PD1
BCA101
BCA101 + Anti-PD1
Tumor Volume (mm3)
Days
*

Conclusions

These results support clinical development of BCA101 as a targeted immunotherapy with potential to induce improved anti-tumor response with a wider therapeutic window, either as a monotherapy or in combination with immune checkpoint blockade therapy.

BCA101 is currently being evaluated in dose escalation studies as a monotherapy and in combination with pembrolizumab (anti-PD-1) - BCA101X1101 (NCT04429542)

Acknowledgements: The authors would like to thank Avanish Varshney, Rachel Salazar and Claire Mazumdar from Bicara Therapeutics for their contributions.