Combination with BCA101 improves the efficacy of KRAS-G12C inhibitor and overcomes G12C inhibitor-induced resistance in lung and colon cancer cells

BICARA therapeutics

sitc Society for Immunotherapy of Cancer

#1426

Reshmi Nair[1]*, Veena Somasundaram[1], Shiv Ram Krishn[1], Rachel Salazar[2], Pradip Nair[1#]

Author Affiliations: [1]Syngene International Limited, Bengaluru, India, and [2]Bicara Therapeutics, Boston, MA, United States.

*Presenting Author and #Corresponding Author

Background

- ❑ Kirsten ras oncogene homolog (KRAS)-G12C allele-specific mutations are majorly observed in about 14% of lung cancer and about 3% of colorectal cancers. EGFR targeting therapies are not efficacious in tumors with G12C mutation. KRAS-G12C inhibitors(G12Ci) have had modest benefits in clinical studies. Initially, G12C mutated tumors respond to the treatment with G12Ci. However, the remissions observed are not sustained and most patients relapse after therapy[1].
- ❑ The mechanisms proposed for G12Ci relapse include upregulation of alternate Receptor Tyrosine Kinase (RTK) pathways and Epithelial to Mesenchymal Transition (EMT) signature triggered by enhanced TGF-β expression within the tumor microenvironment (TME)[1,2,3].
- ❑ In the present study, we evaluated whether BCA101, a bispecific antibody consisting of TGFβRII-ECD(trap) fused to a heavy chain of an anti-EGFR antibody, abrogates the KRAS-G12Ci induced resistance in lung and colon cancer preclinical models by targeting EGFR and sequestering TGF-β within tumors.

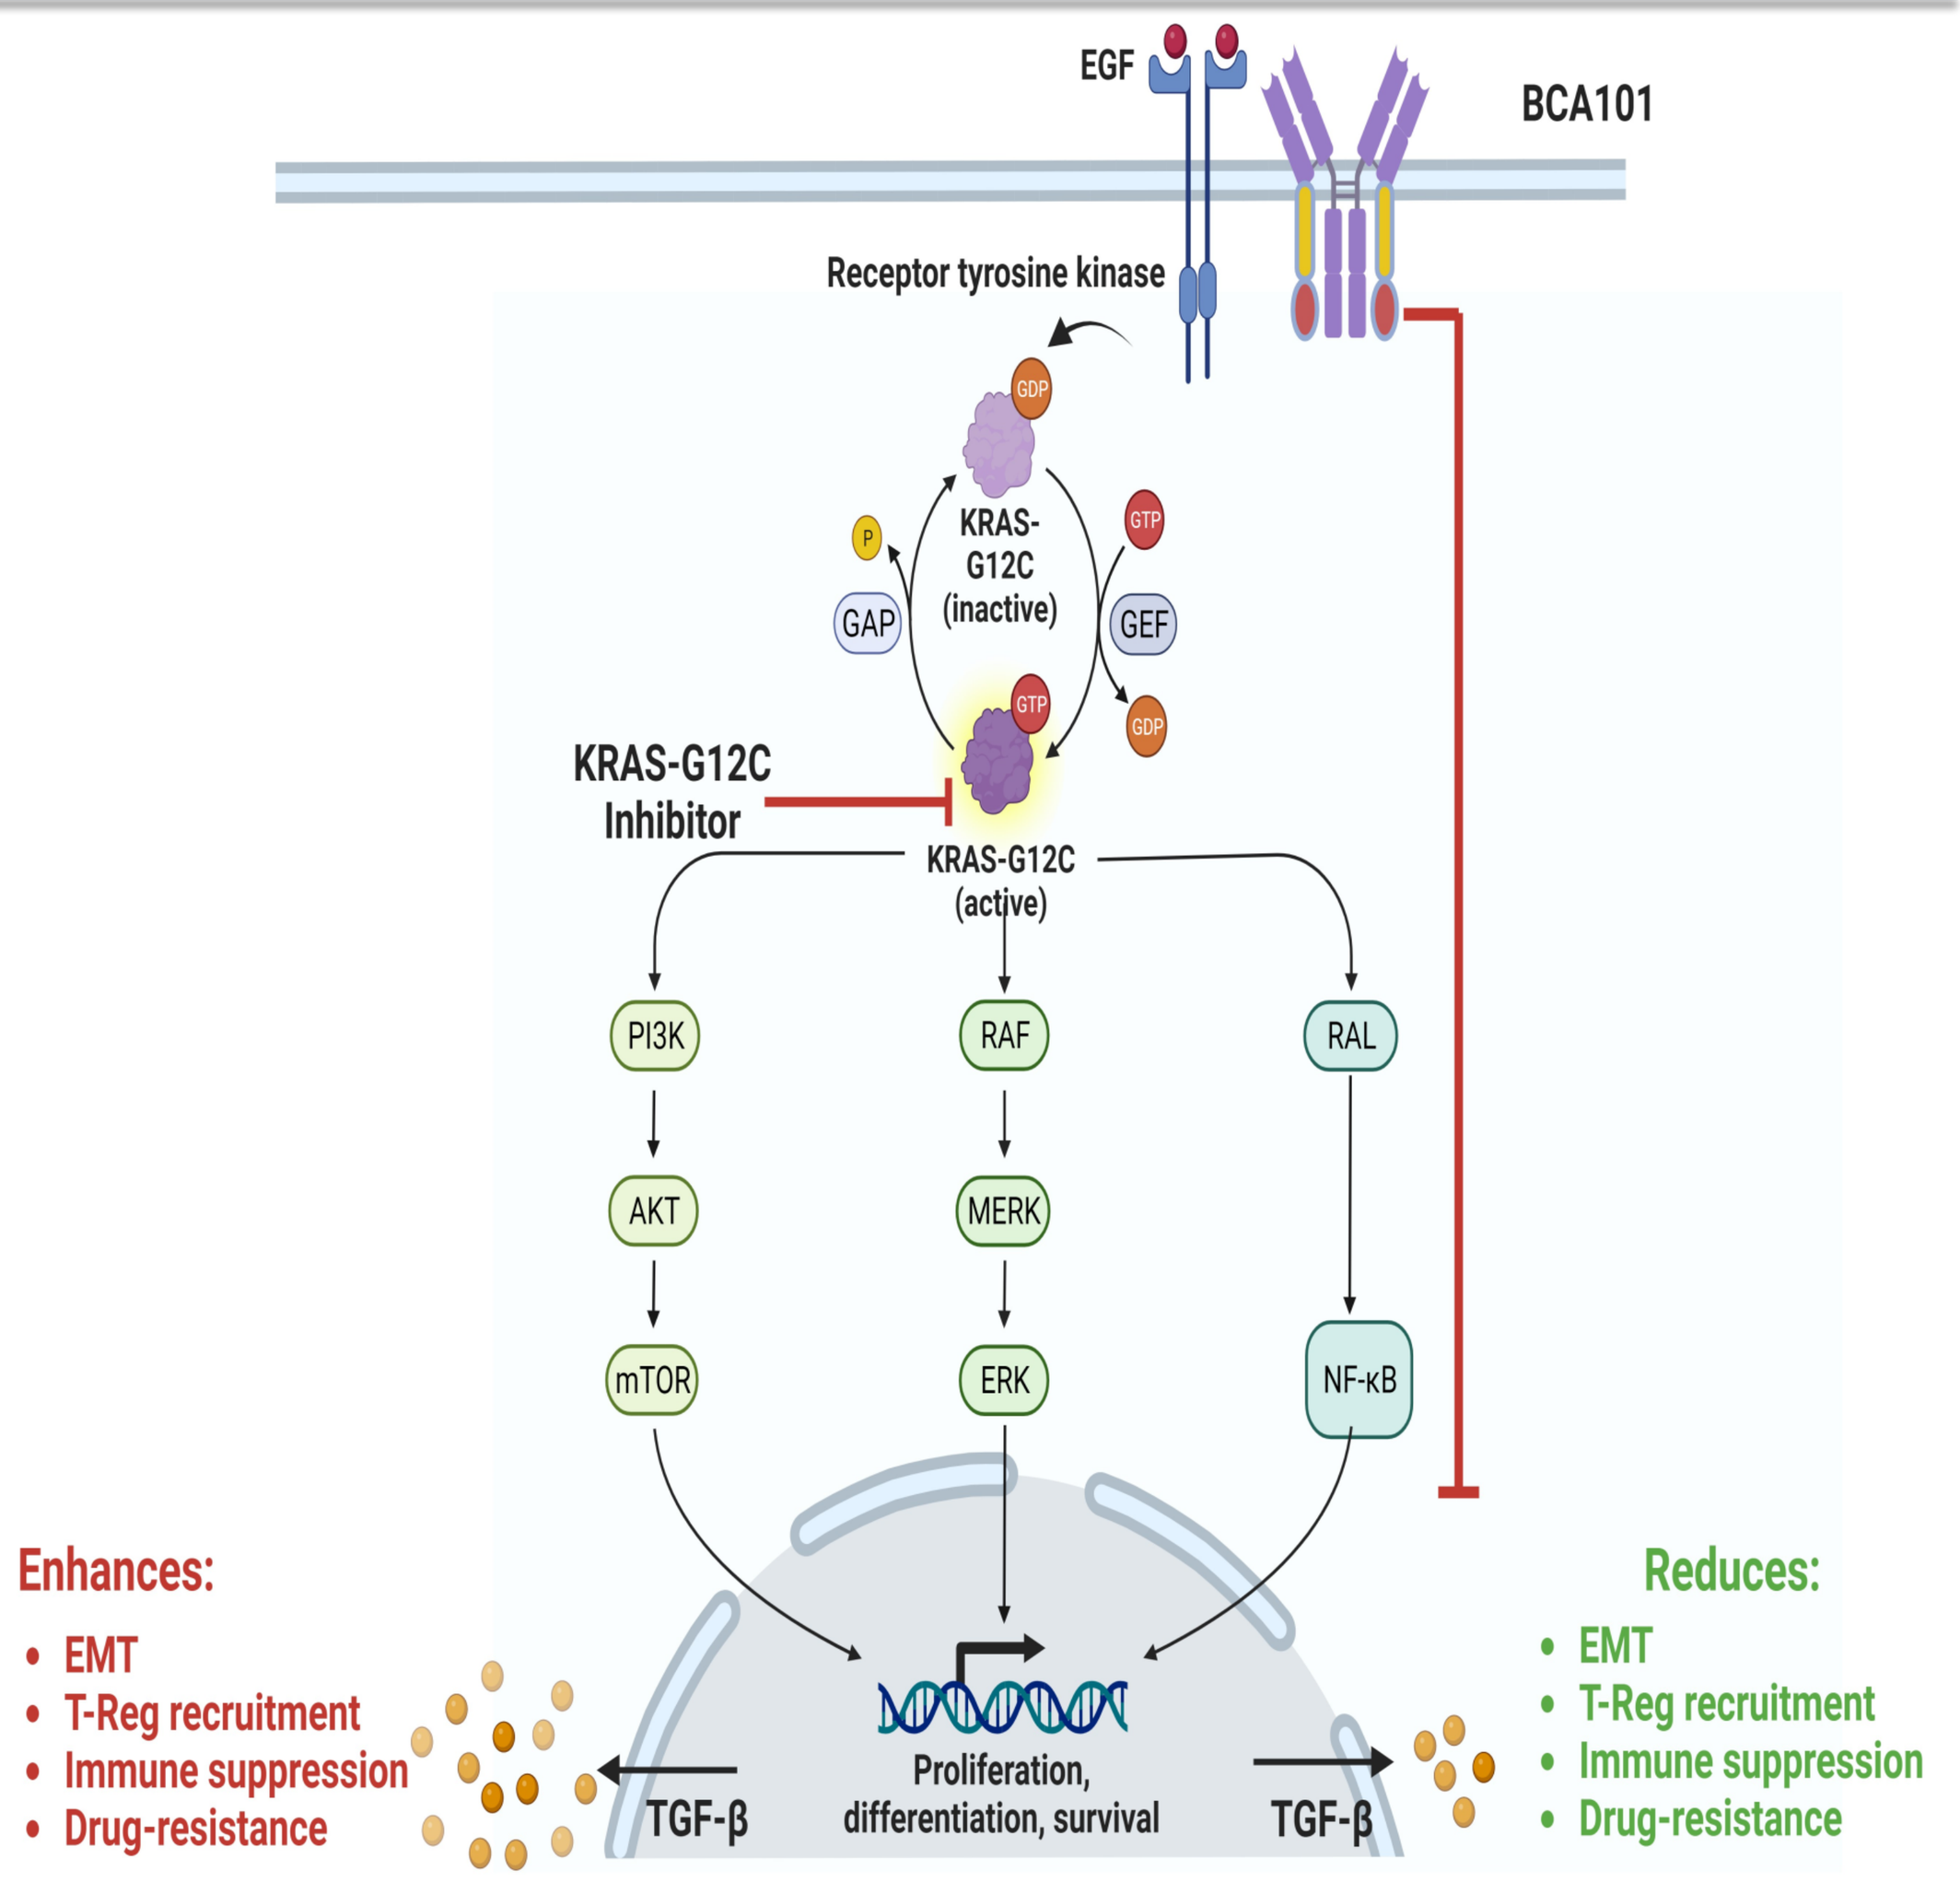
EGF
BCA101
Receptor tyrosine kinase
KRAS-G12C (inactive)
GAP
GEF
KRAS-G12C Inhibitor
KRAS-G12C (active)
PI3K
AKT
mTOR
RAF
MERK
ERK
RAL
NF-κB
Enhances:
EMT
T-Reg recruitment
Immune suppression
Drug-resistance
TGF-β
Proliferation, differentiation, survival
TGF-β
Reduces:
EMT
T-Reg recruitment
Immune suppression
Drug-resistance

Figure1: EGFR signaling and KRAS-G12Ci induced resistance mechanism: Upon binding with the ligand, EGFR signaling activates KRAS and downstream signaling PI3K/RAF/NFκB pathways. The signaling cascade leads to proliferation, differentiation and survival of cancer cells. KRAS-G12Ci inhibits the growth of G12C mutated tumors but acquires resistance due to upregulation of TGF-β as one of the mechanisms. Combination of KRAS-G12Ci with BCA101 traps TGF-β upregulated in G12Ci resistant tumors, improves efficacy and overcomes drug mediated resistance. Figure made using BioRender.

EGFR and TGF-β expression in KRAS-G12C mutated lung and colorectal cancer cell lines

A

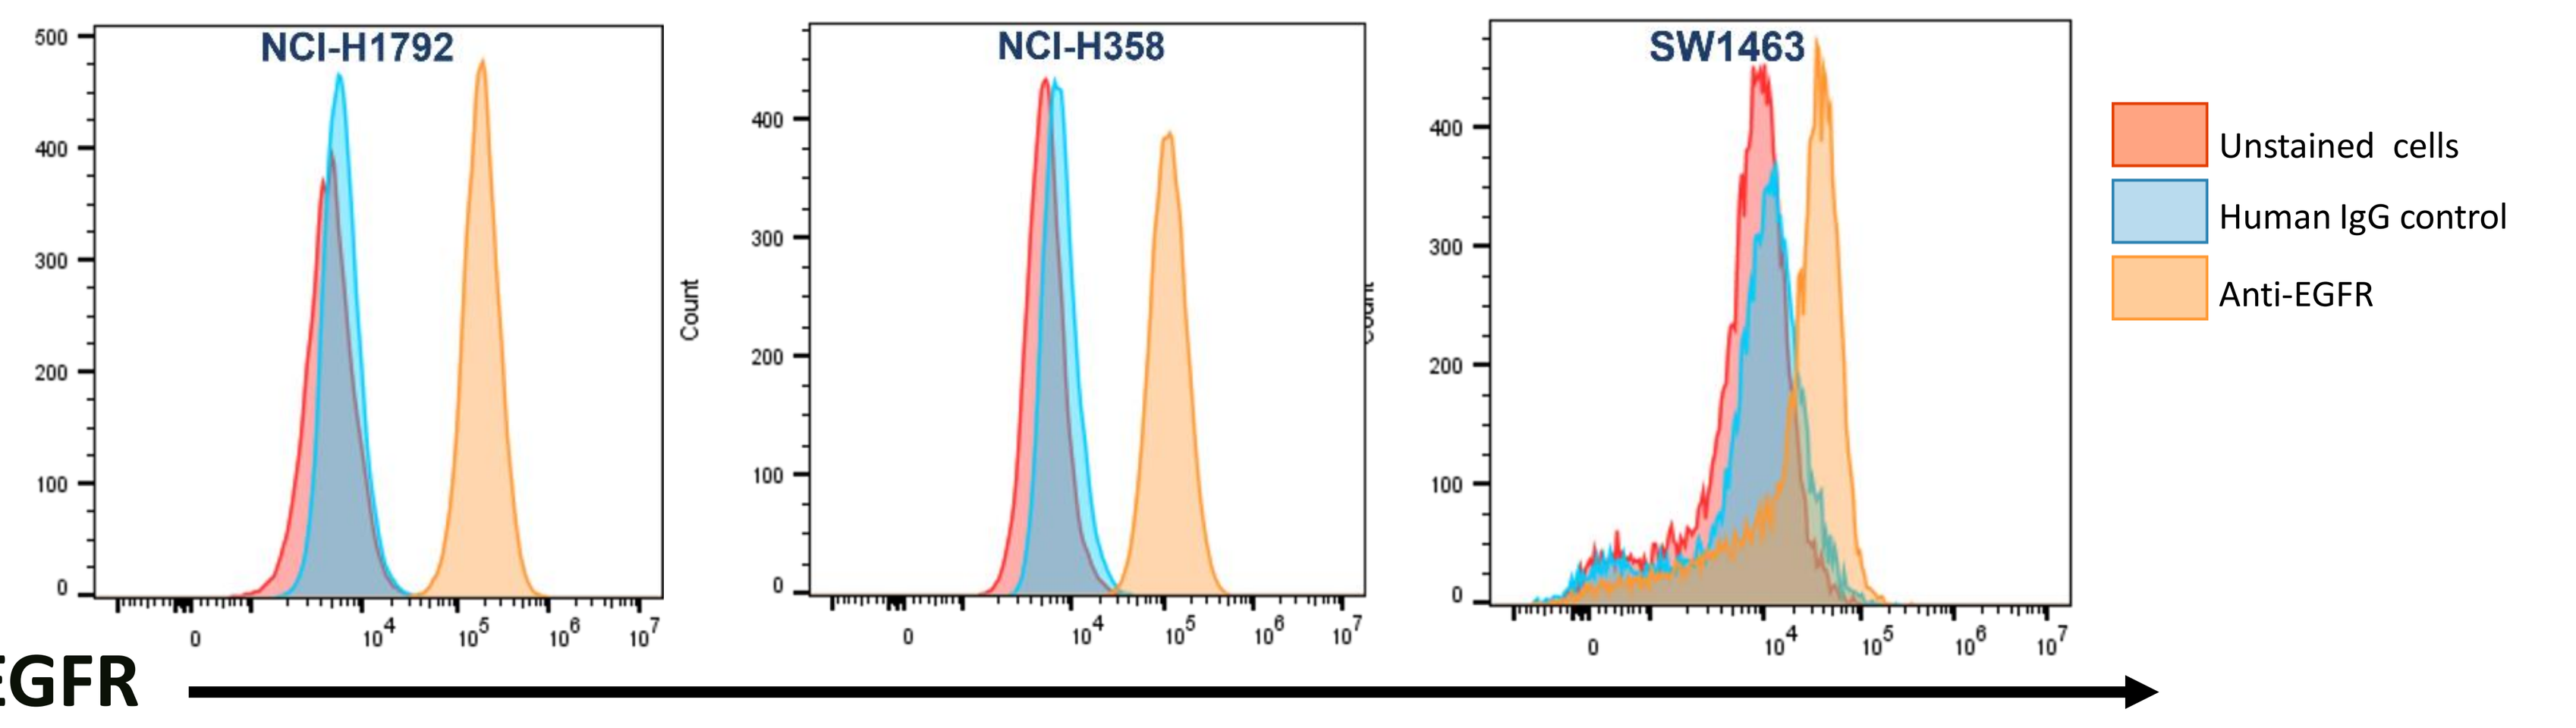
NCI-H1792
NCI-H358
SW1463
Count
Unstained cells
Human IgG control
Anti-EGFR
EGFR

B

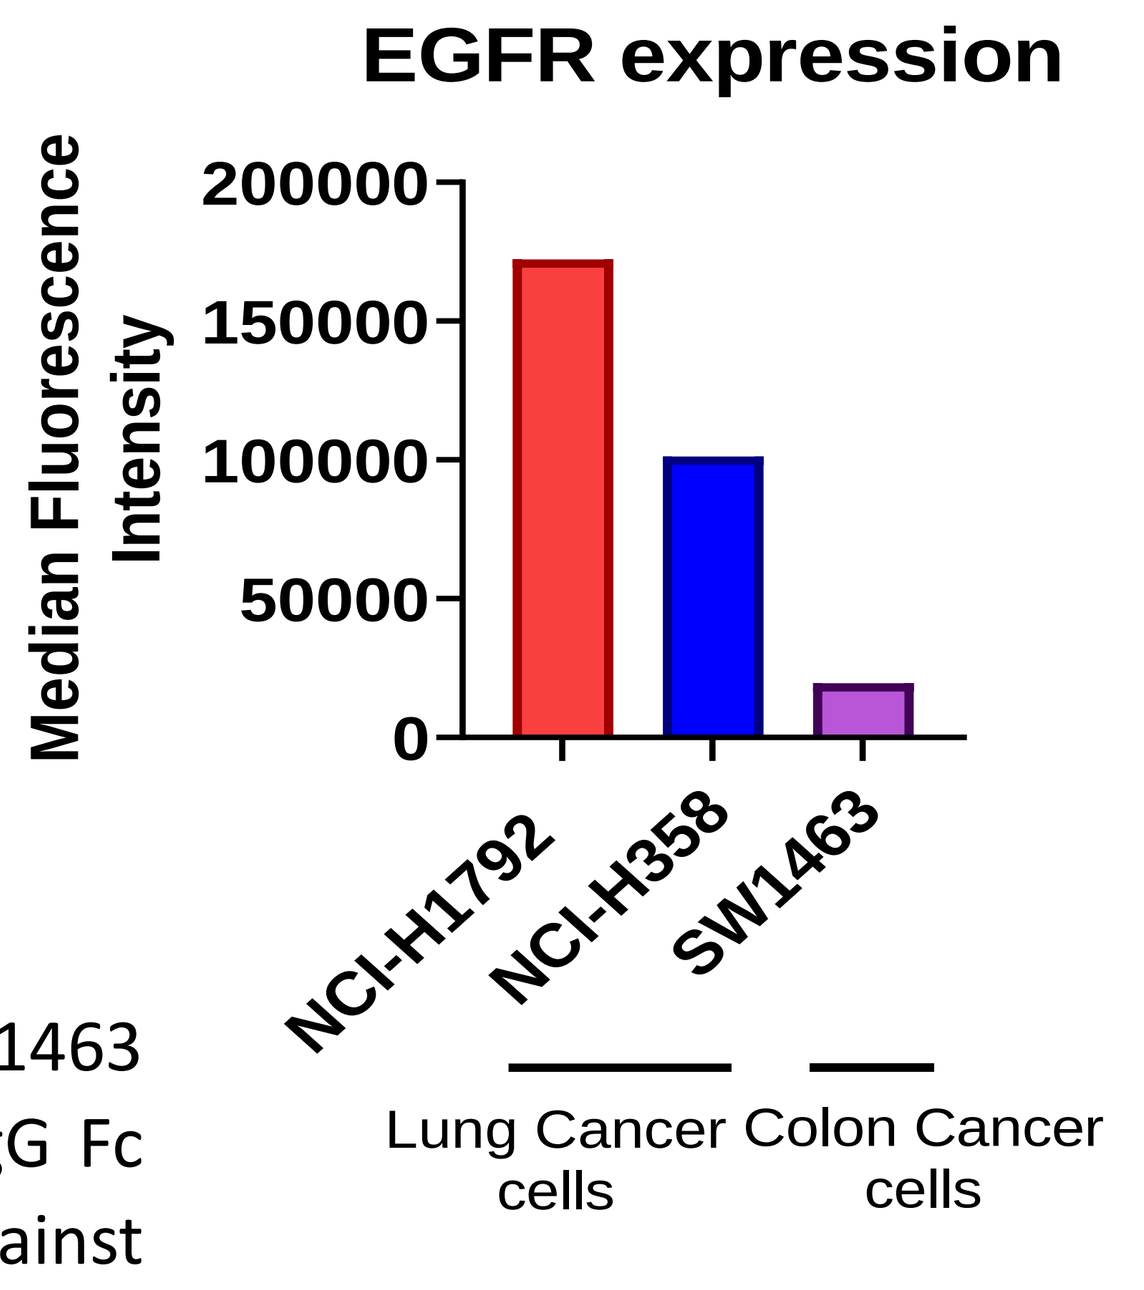
EGFR expression
Median Fluorescence Intensity
200000
150000
100000
50000
0
NCI-H1792
NCI-H358
SW1463
Lung Cancer cells
Colon Cancer cells

C

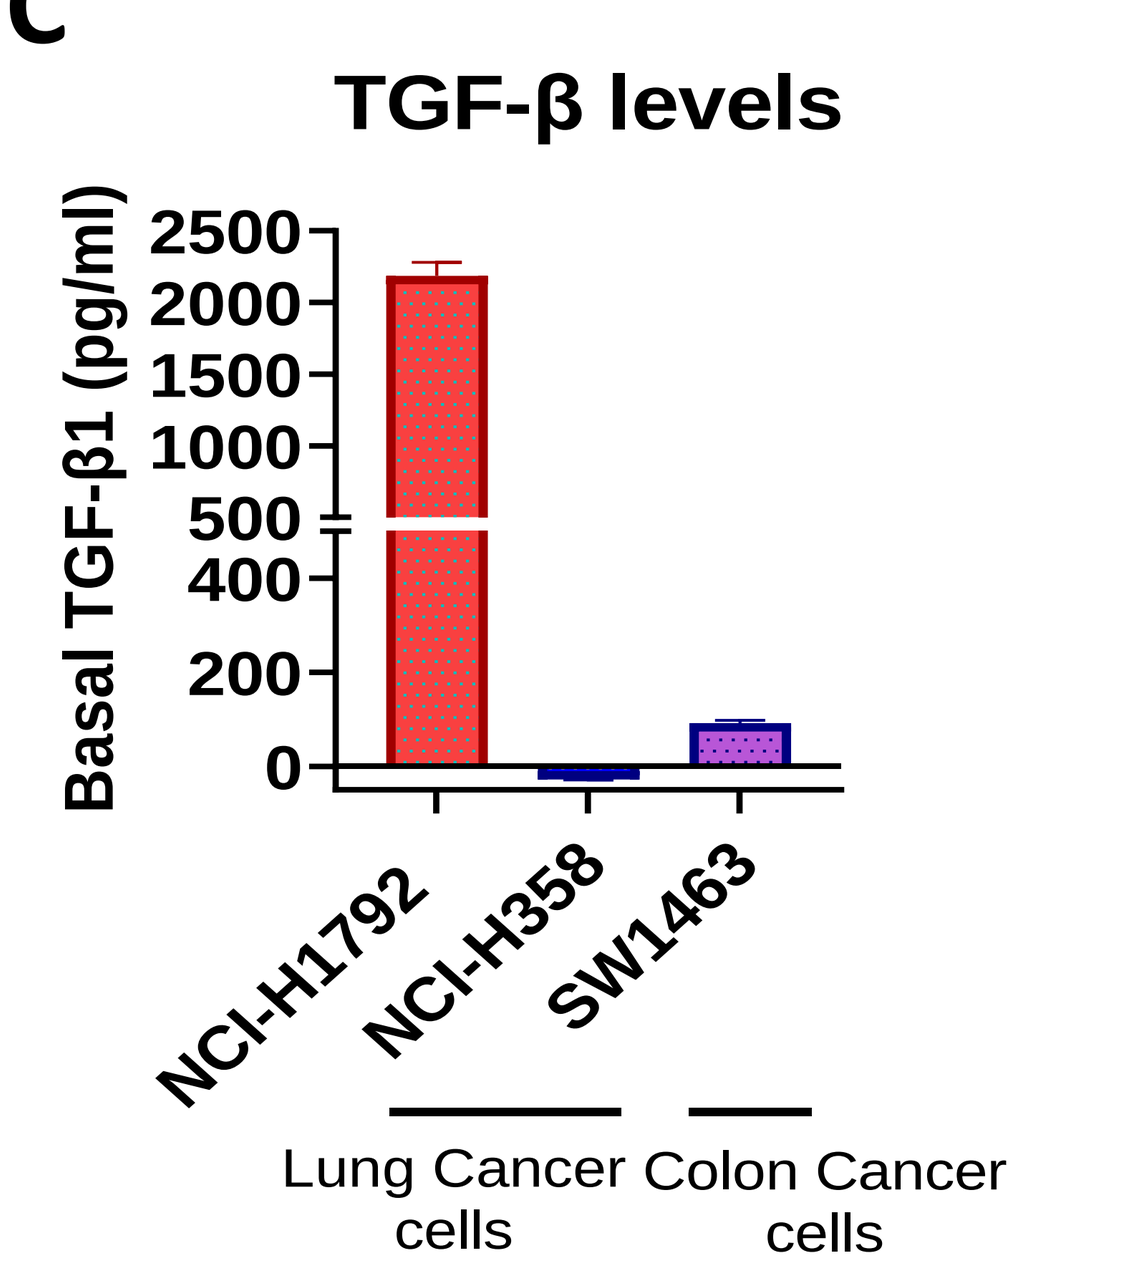
TGF-β levels
Basal TGF-β1 (pg/ml)
2500
2000
1500
1000
500
400
200
0
NCI-H1792
NCI-H358
SW1463
Lung Cancer cells
Colon Cancer cells

Figure2: EGFR expression and basal TGF-β secretion level in KRAS-G12C mutated lung and colon cancer cells:

(A) EGFR expression evaluated by flow cytometry in NCI-H1792(G12C homozygous), NCI-H358(G12C heterozygous), and SW1463 (G12C homozygous) cells. The cells were stained with anti EGFR antibody and detected using FITC labelled anti-human IgG Fc secondary antibody. Shift in Median Fluorescence Intensity (MFI) showed EGFR expression (Orange) and was compared against controls: Human IgG (Blue) and unstained cells (Red). **(B)** Comparative MFI of EGFR expression across the cell lines. **(C)** 0.5X10^6 cells were seeded and the supernatant was collected after 72h. TGF-β secretion was evaluated using ELISA.

Combination with BCA101 improves response with KRAS-G12Ci and overcomes acquired resistance to KRAS-G12Ci in lung cancer cells

A

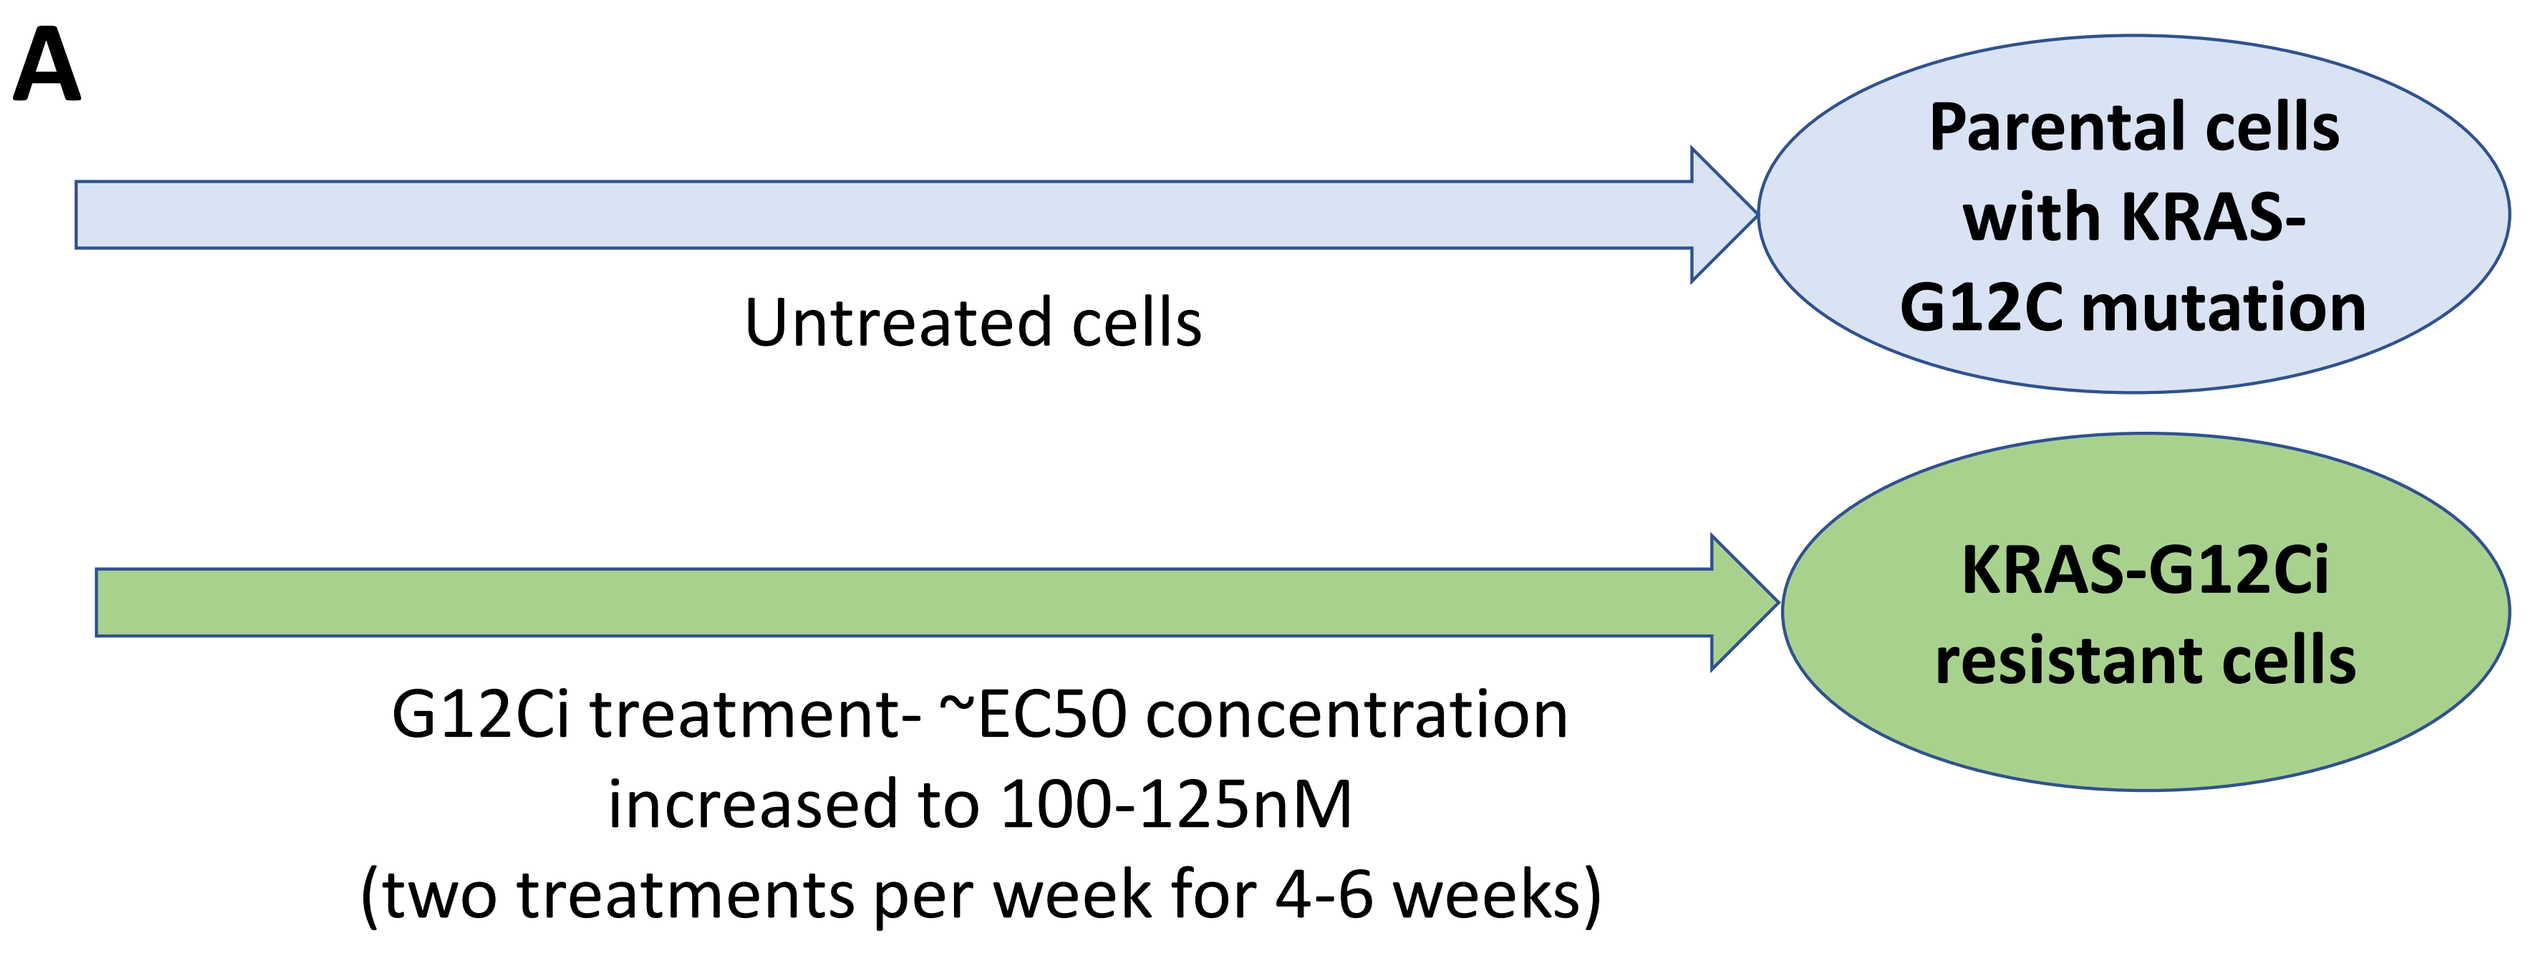
Untreated cells
Parental cells with KRAS-G12C mutation
G12Ci treatment- ~EC50 concentration increased to 100-125nM (two treatments per week for 4-6 weeks)
KRAS-G12Ci resistant cells

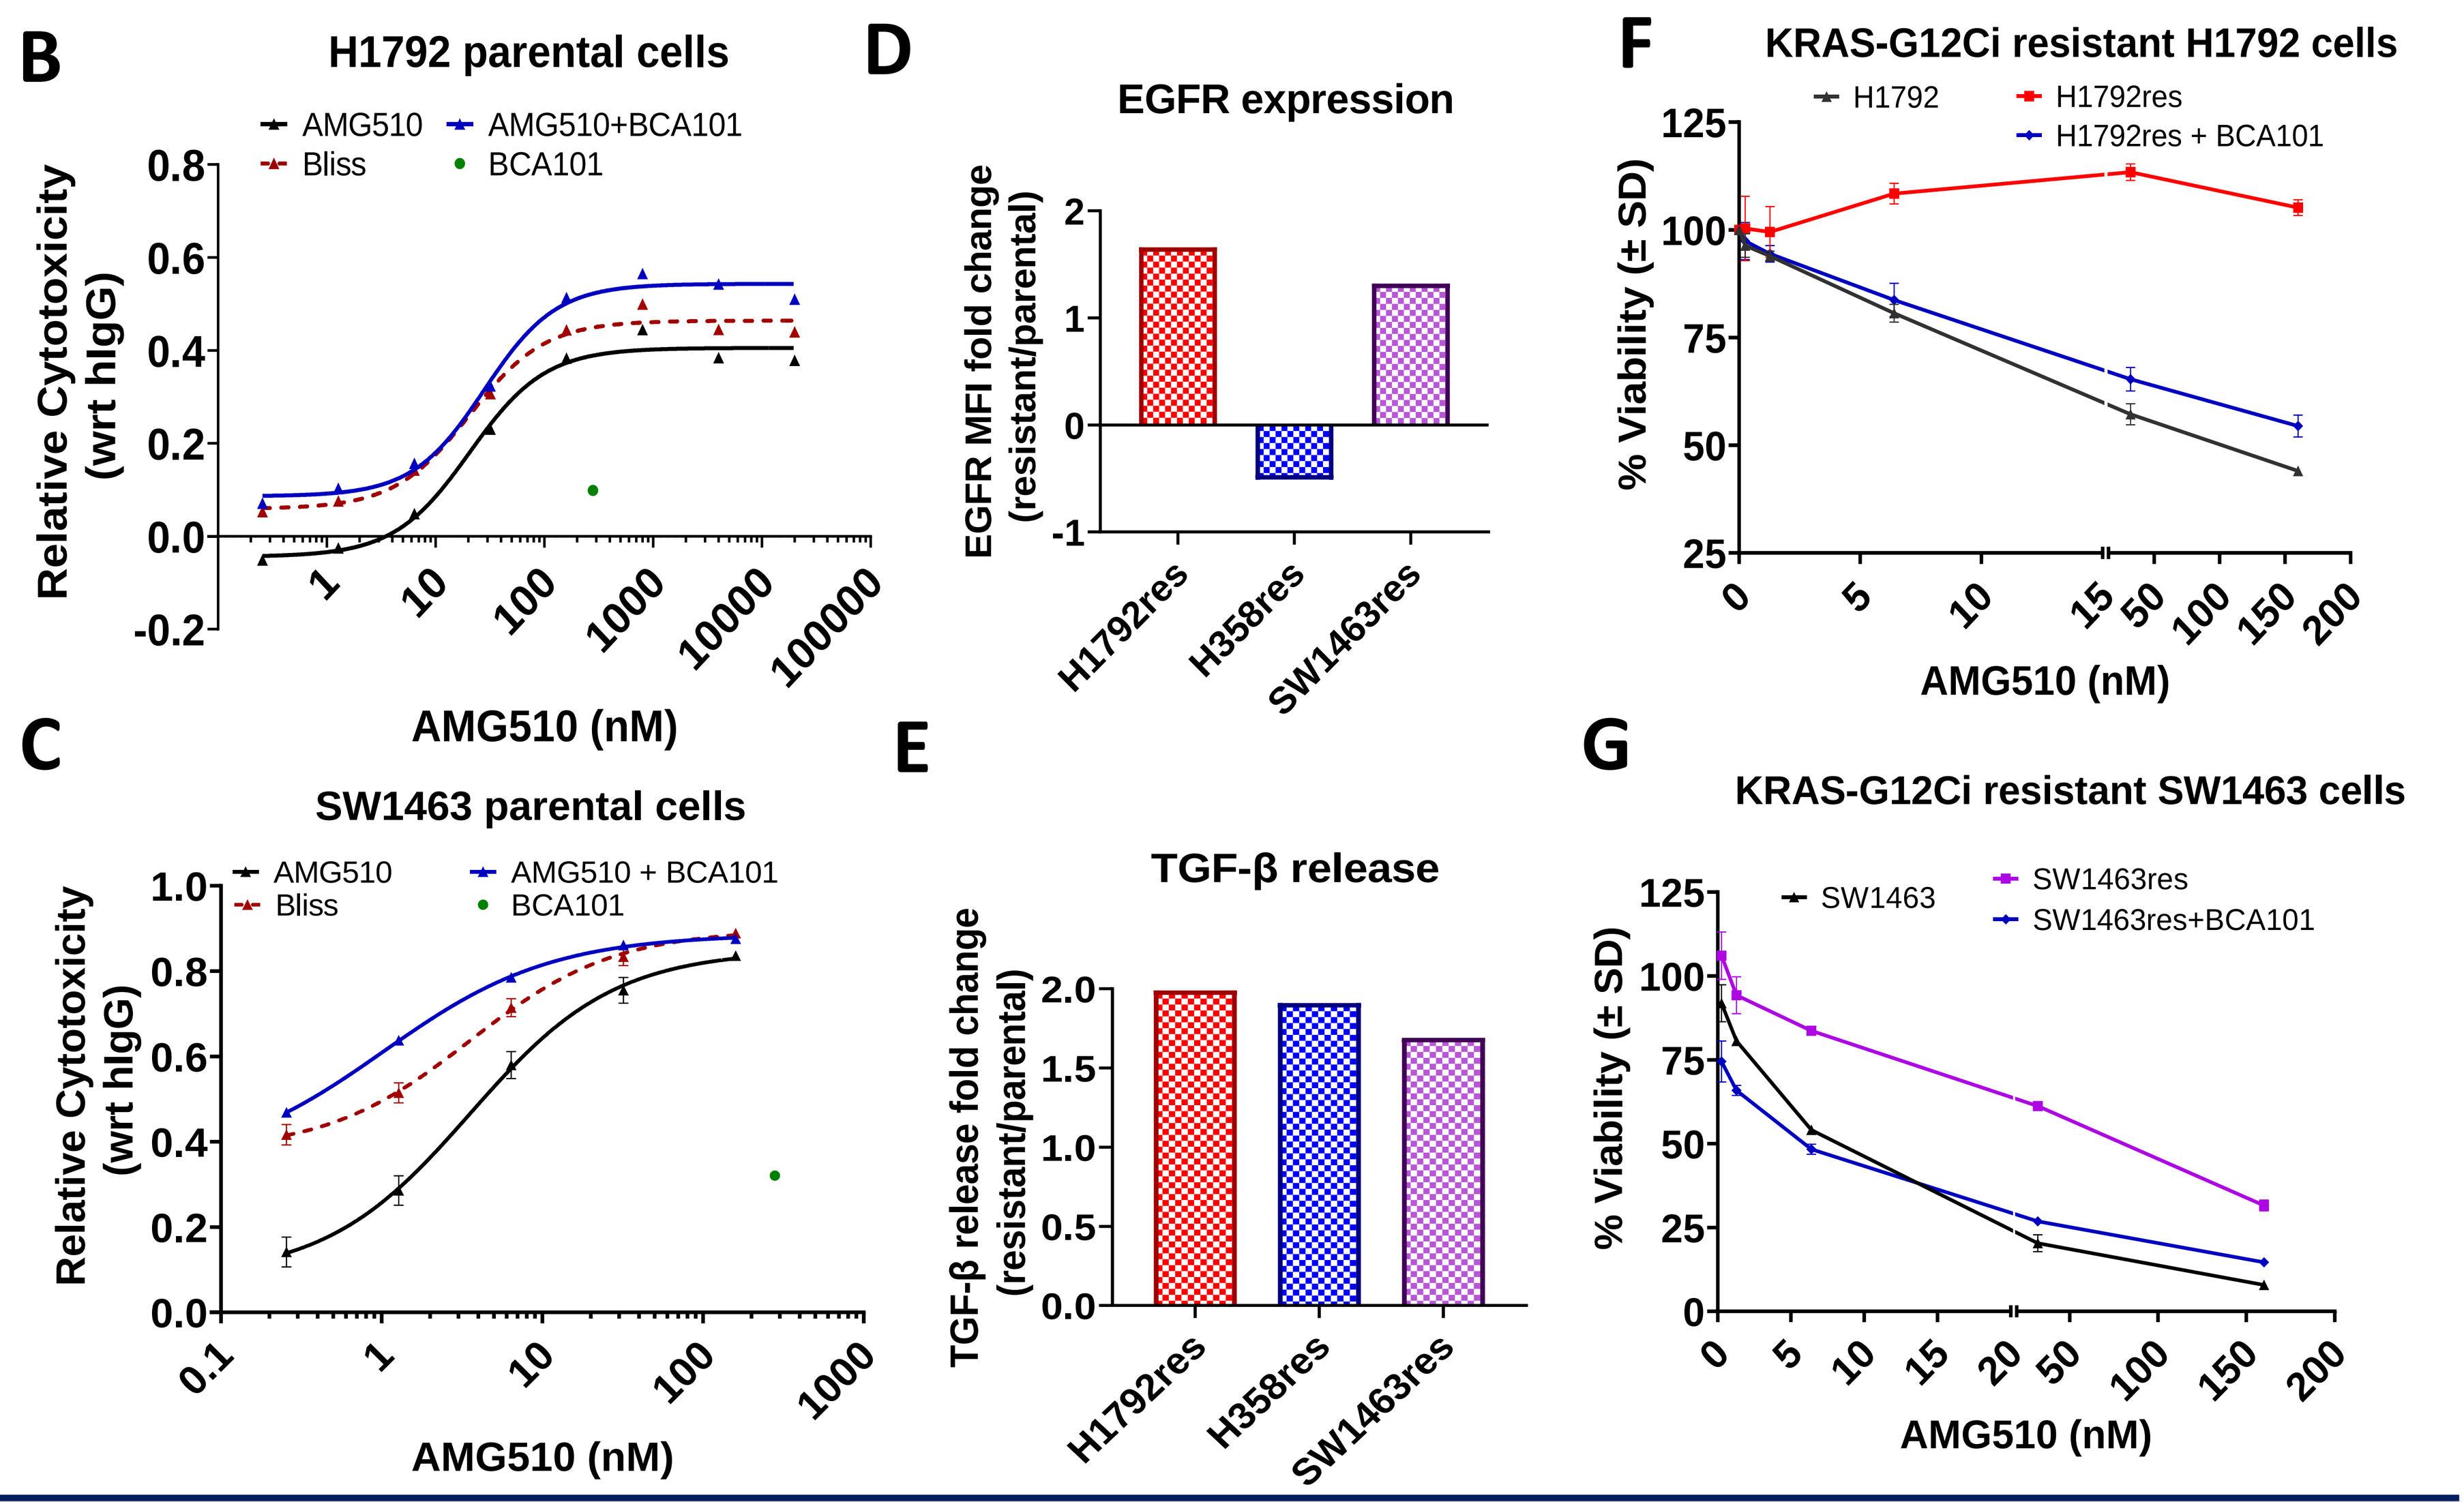
B
H1792 parental cells
AMG510
AMG510+BCA101
Bliss
BCA101
Relative Cytotoxicity (wrt hIgG)
AMG510 (nM)
C
SW1463 parental cells
AMG510
AMG510 + BCA101
Bliss
BCA101
Relative Cytotoxicity (wrt hIgG)
AMG510 (nM)
D
EGFR expression
EGFR MFI fold change (resistant/parental)
H1792res
H358res
SW1463res
E
TGF-β release
TGF-β release fold change (resistant/parental)
H1792res
H358res
SW1463res
F
KRAS-G12Ci resistant H1792 cells
H1792
H1792res
H1792res + BCA101
% Viability (± SD)
AMG510 (nM)
G
KRAS-G12Ci resistant SW1463 cells
SW1463
SW1463res
SW1463res+BCA101
% Viability (± SD)
AMG510 (nM)

Figure3: Schematic for the generation of KRAS-G12Ci resistant cancer cells and features of resistant cells: (A) Flowchart shows generation of KRAS-G12Ci resistant cells by prolonged exposure to KRA-G12Ci over 4-6 weeks. **(B,C)** Dose response curve of cytotoxicity induced by KRAS-G12Ci (AMG510) singly or in combination with BCA101 after 72h treatment (B) or 120h treatment (C). The combination shows synergistic effect as determined by Bliss analysis (*Bliss = Cytotoxicity A+B–(AXB)*; Experimental Cytotoxicity(A+B) > Bliss = Synergism {A:AMG510, B: BCA101}. **(D,E)** H1792 and SW1463 resistant cells (H1792res and SW1463res respectively), show an increase in EGFR surface expression and TGF-β release**,** while H358 show increase in TGF-β release but surface EGFR expression is reduced in resistant cells**. (F,G)** H1792res and SW1463res cells remain susceptible to AMG510+BCA101 combination.

Combination of BCA101 with AMG510 is efficacious over AMG510 single treatment and reduces growth of H358 xenograft tumors

A

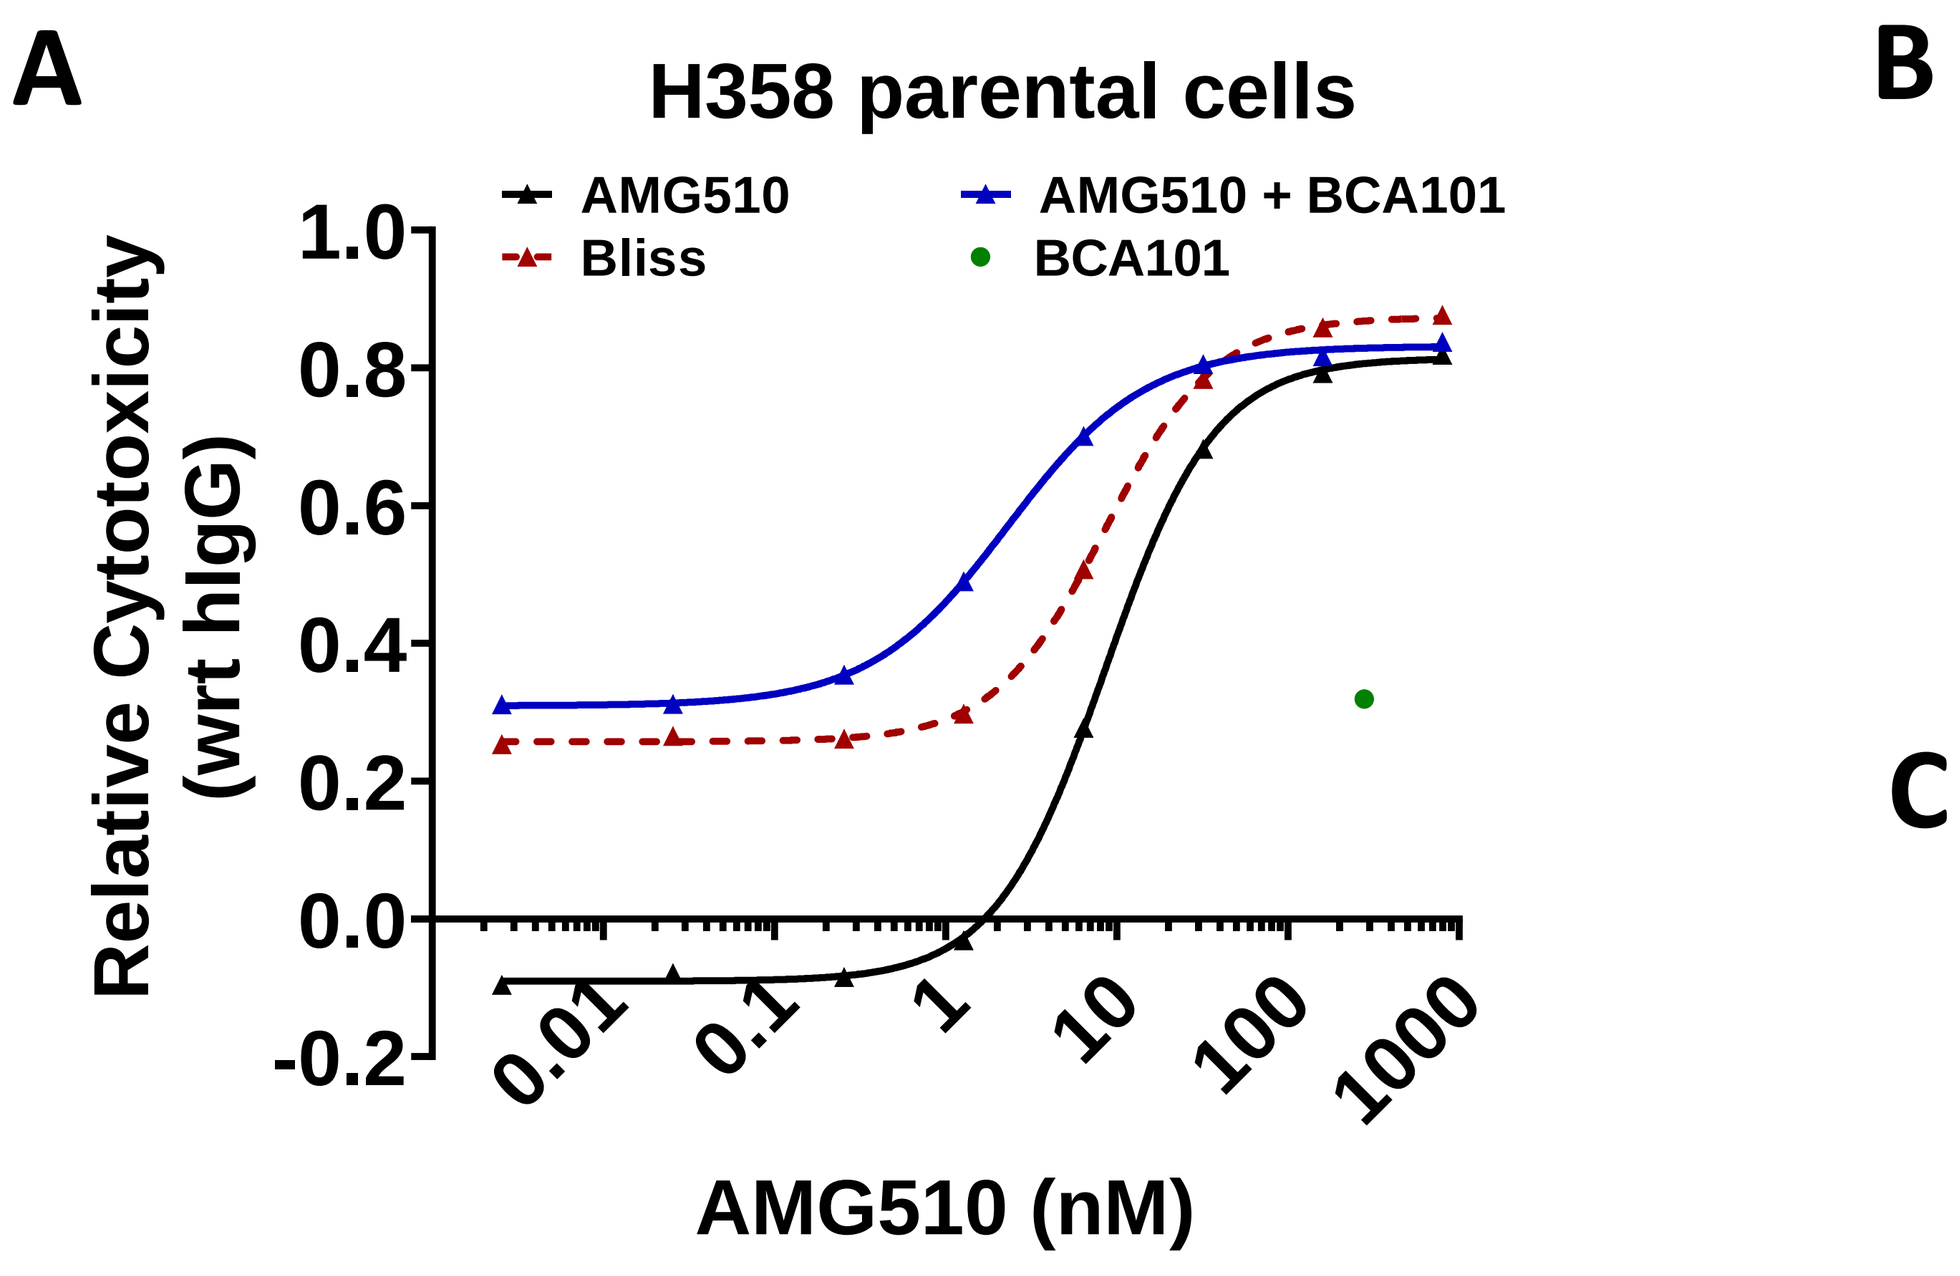
H358 parental cells
AMG510
AMG510 + BCA101
Bliss
BCA101
Relative Cytotoxicity (wrt hIgG)
AMG510 (nM)

B

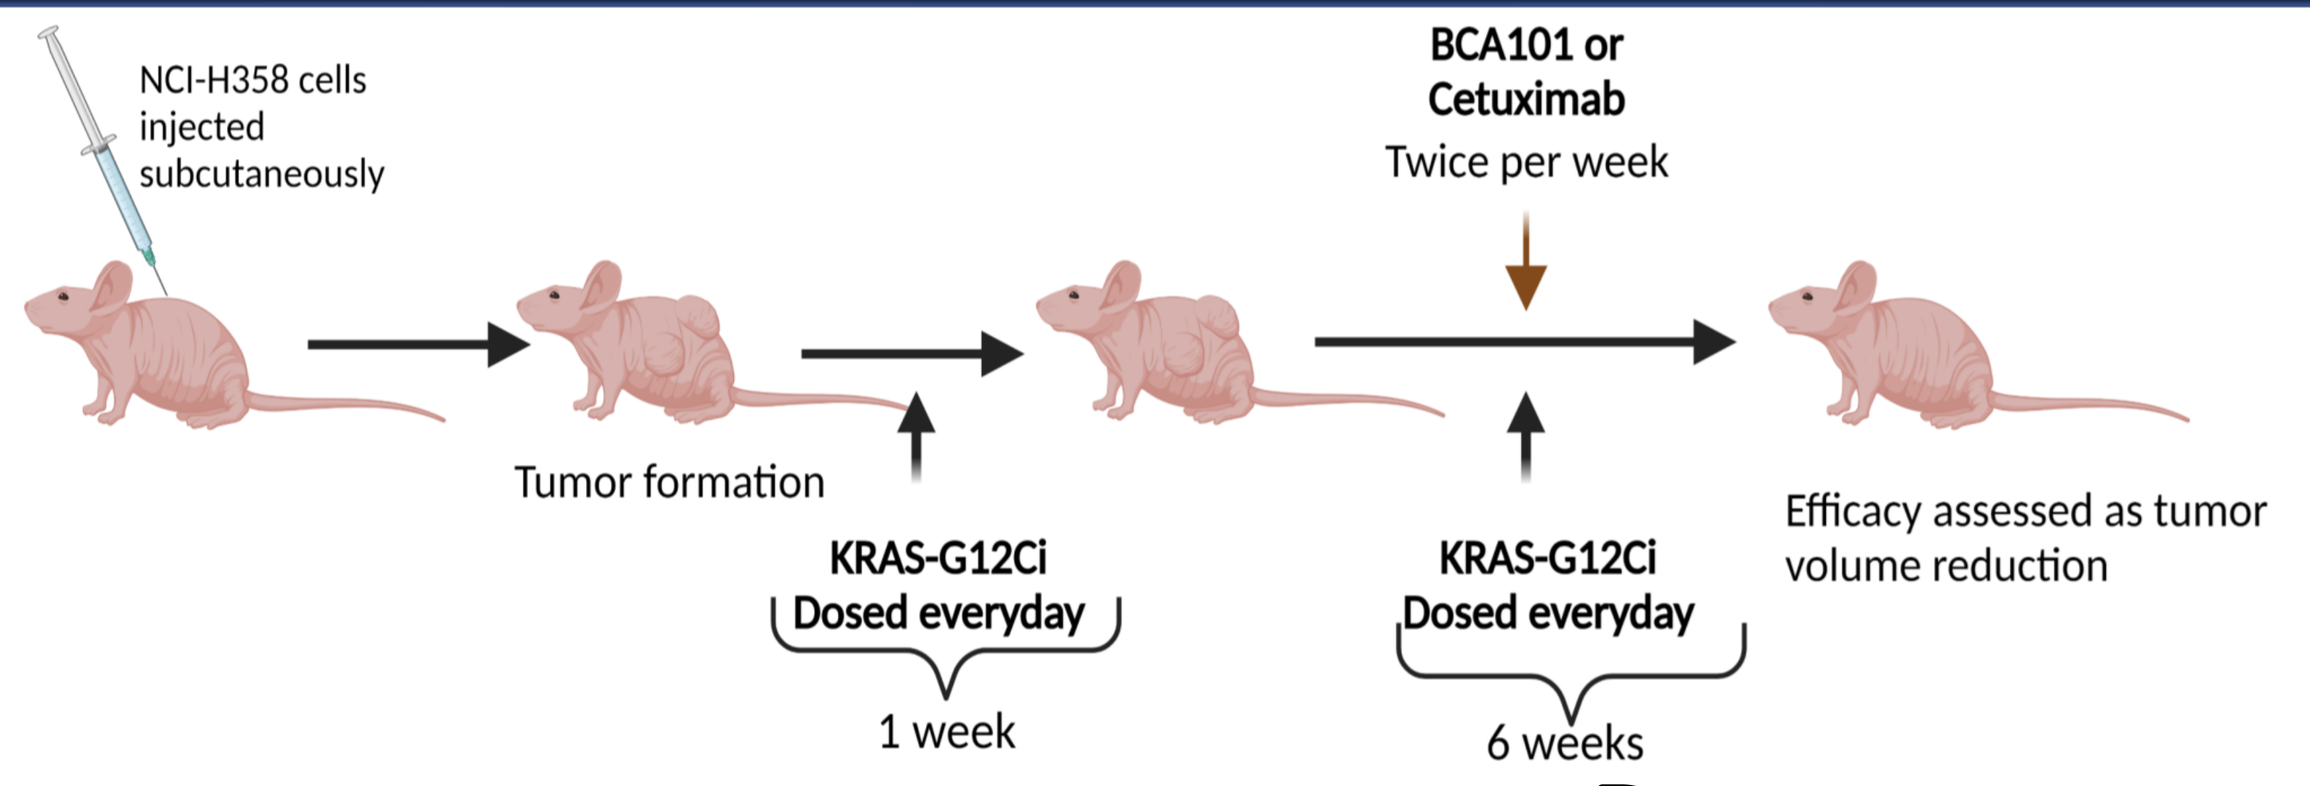
NCI-H358 cells injected subcutaneously
Tumor formation
KRAS-G12Ci Dosed everyday
1 week
BCA101 or Cetuximab
Twice per week
KRAS-G12Ci Dosed everyday
6 weeks
Efficacy assessed as tumor volume reduction

C D

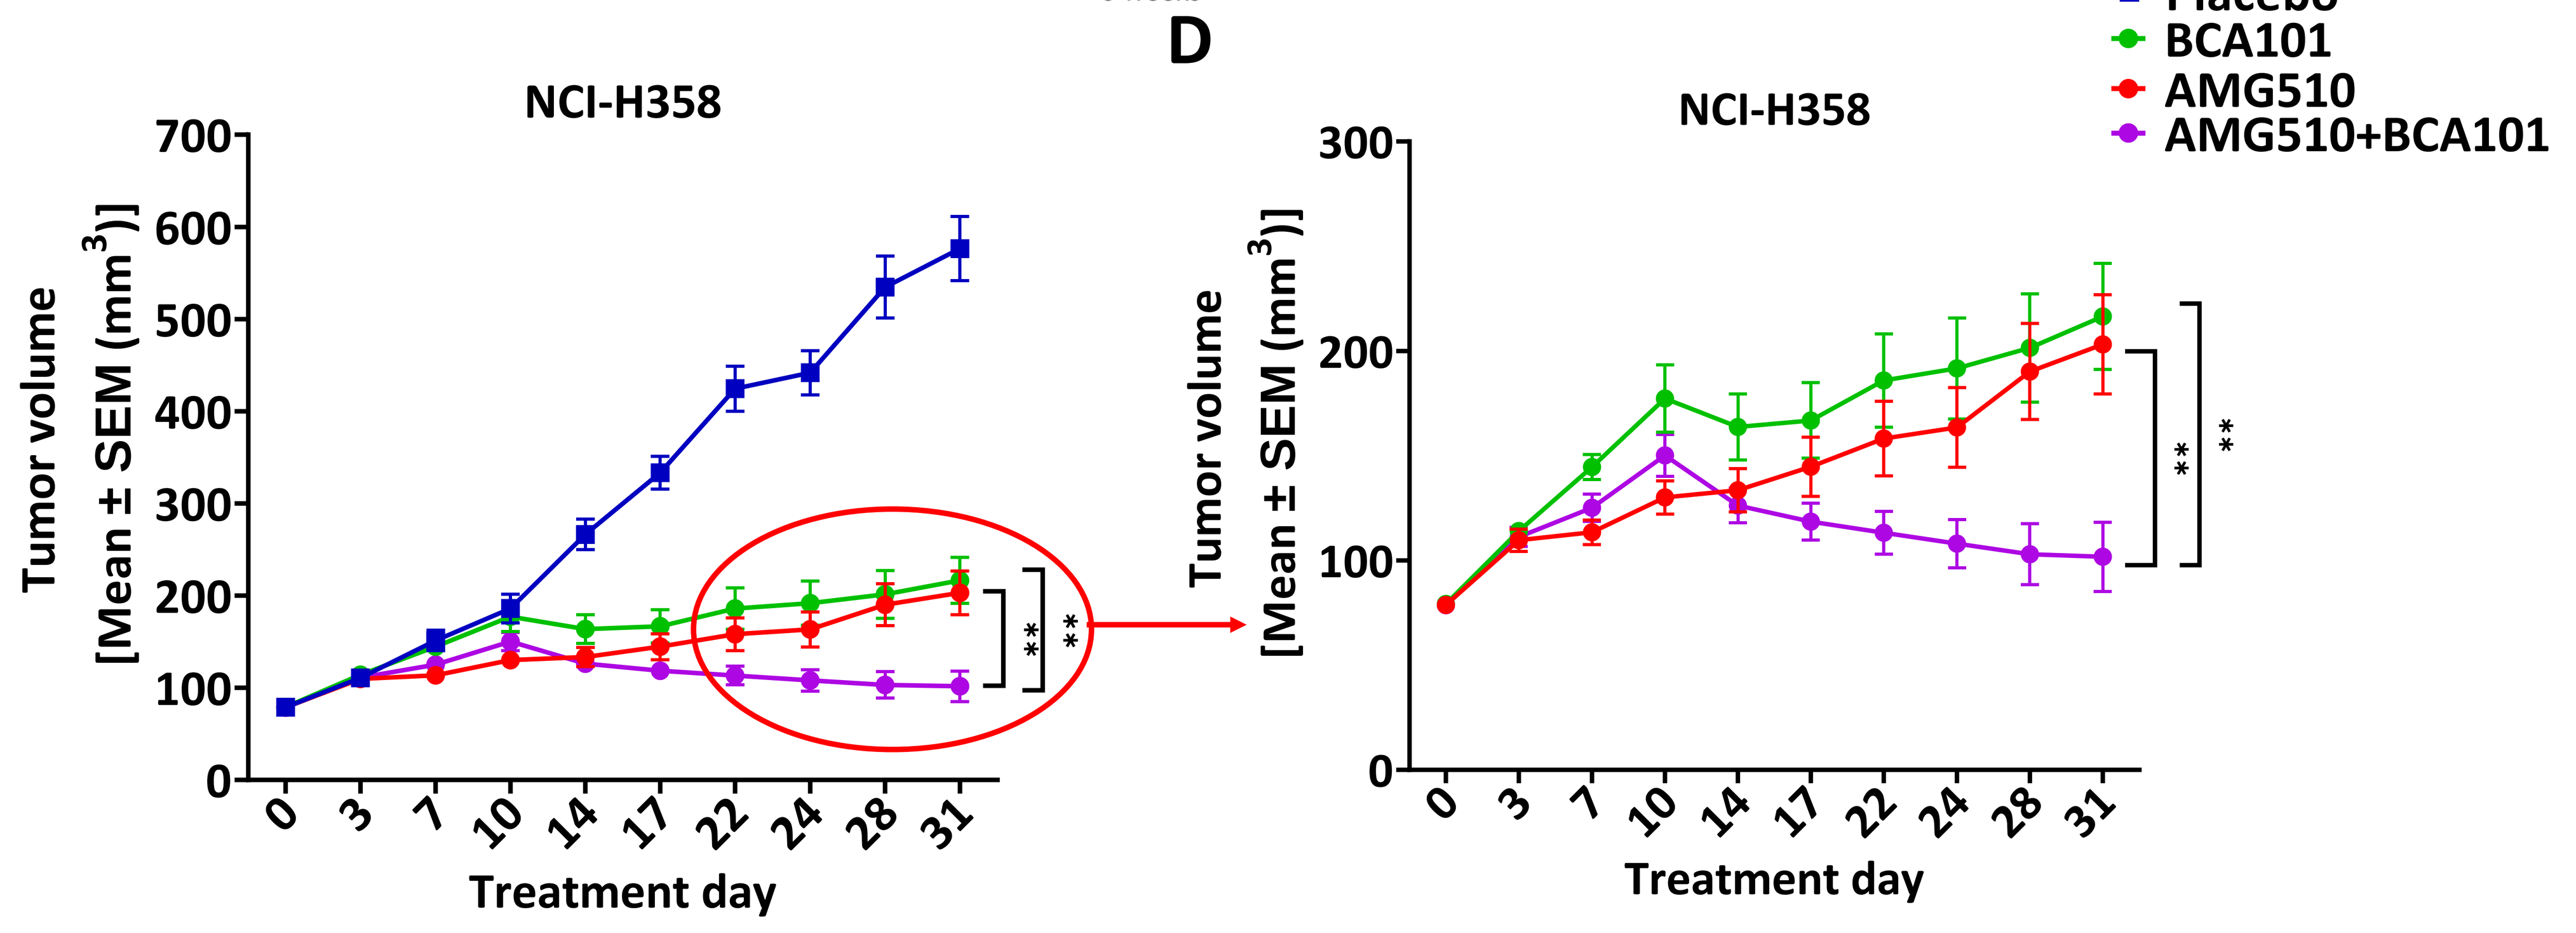
NCI-H358
Tumor volume [Mean ± SEM (mm3)]
Treatment day
NCI-H358
Placebo
BCA101
AMG510
AMG510+BCA101
**

Figure 4: ***In vivo*** **efficacy study :(A)** Dose response curve of cytotoxicity induced by KRAS-G12Ci (AMG510) singly or in combination with BCA101 after 72h treatment **(B)** Schematic of the dosing strategy in athymic nude mice**. (C)** NCI-H358 xenograft tumors growth measured in nude mice treated with either placebo, or BCA101 alone (5mg/kg), AMG510 alone (5mg/kg), and AMG510 (5mg/kg) + BCA101 (5mg/kg), on days as shown in the figure. Data are mean tumor volume ±SEM (mm^3), n = 10 mice per group. **(D)** Enlarged image excluding placebo group.

Conclusions

- ❑ *This data confirms that, in the cell lines tested, prolonged treatment with G12Ci increases TGF-β, which is reported as one of the resistance mechanisms.*
- ❑ *BCA101 in combination with G12Ci is more efficacious than G12Ci alone. G12Ci resistant cell lines remain susceptible to this combination treatment.*
- ❑ *The data suggests that basal expression of EGFR and TGF-β is critical to determine the efficacy of combination of G12Ci with BCA101. Patient stratification based on expression of EGFR and TGF-β can be beneficial for treatment of patients with KRAS-G12C mutated tumors.*
- ❑ *BCA101 has the potential to increase the efficacy of G12Ci along with the ability to mitigate G12Ci-induced acquired resistance in lung and colon cancer derived cell lines. This study provides a rationale that combination of BCA101 with G12Ci in G12C mutant lung or colon cancer patients has the potential to show good clinical efficacy.*

Acknowledgements

The authors would like to thank Syngene International Limited for their vivarium facility and performing the animal experiments.

References

1. Adachi et.al., Epithelial-to-Mesenchymal Transition is a cause of both intrinsic and acquired resistance to KRAS-G12C inhibitor in KRAS-G12C–mutant non–small cell lung cancer, Clin Cancer Res, 2020.
2. Huang et.al., KRAS mutation: from undruggable to druggable in cancer, Signal Transduction and Targeted Therapy, 2021.
3. Ryan et.al., KRASG12C independent feedback activation of wildtype RAS constrains KRASG12C inhibitor efficacy, Cell Reports, 2022.