1180

Dual targeting of TGF-β and EGFR with ficerafusp alfa shows superior antitumor activity over cetuximab in KRAS/BRAF wild-type MSS colorectal cancer cell lines

Anshu Kuriakose,[1] Reshmi Nair,[1] Pradip Nair,[1] Brenda C O'Connell,[2] Rachel Salazar[2]

[1]Syngene International Ltd, Bengaluru, India; [2]Bicara Therapeutics Inc., Boston, MA, USA

KEY TAKEAWAYS

Dual targeting of EGFR and TGF-β with ficerafusp alfa showed improved tumor cytotoxicity, enhanced expression of proinflammatory factors, and reduced expression of protumor factors compared with cetuximab in KRAS/BRAF wild-type (WT), microsatellite-stable (MSS) colorectal cancer (CRC) cell lines

- In the absence of peripheral blood mononuclear cells (PBMCs), NCI-H508 cells were sensitive to ficerafusp alfa and cetuximab, whereas SNU-503 cells with high basal expression of TGF-β1 were largely resistant
- In the presence of PBMCs, ficerafusp alfa enhanced IFN-γ secretion and the cytolytic activity of PBMCs against both NCI-H508 and SNU-503 cells
- The superior effect of ficerafusp alfa over cetuximab was maintained in the presence of cancer-associated fibroblast (CAF)-conditioned media

The enhanced and durable antitumor effect of ficerafusp alfa in PBMC-tumor coculture assays supports a potential therapeutic advantage of tumor-targeted inhibition of TGF-β in overcoming immune suppression associated with anti-PD-1 and anti-EGFR resistance in MSS CRC

Ficerafusp alfa downregulates VEGF, an important angiogenic factor in both left- and right-sided CRC, when compared with cetuximab, supporting the rationale for exploring ficerafusp alfa regardless of sidedness

Ficerafusp alfa is being explored as monotherapy or in combination with pembrolizumab in patients with RAS/BRAF WT MSS metastatic CRC in a phase 1/1b expansion cohort (NCT04429542)

Figure 1. Schematic representation of the mechanism of action of ficerafusp alfa in the TGF-β1–enriched CRC tumor microenvironment (TME).

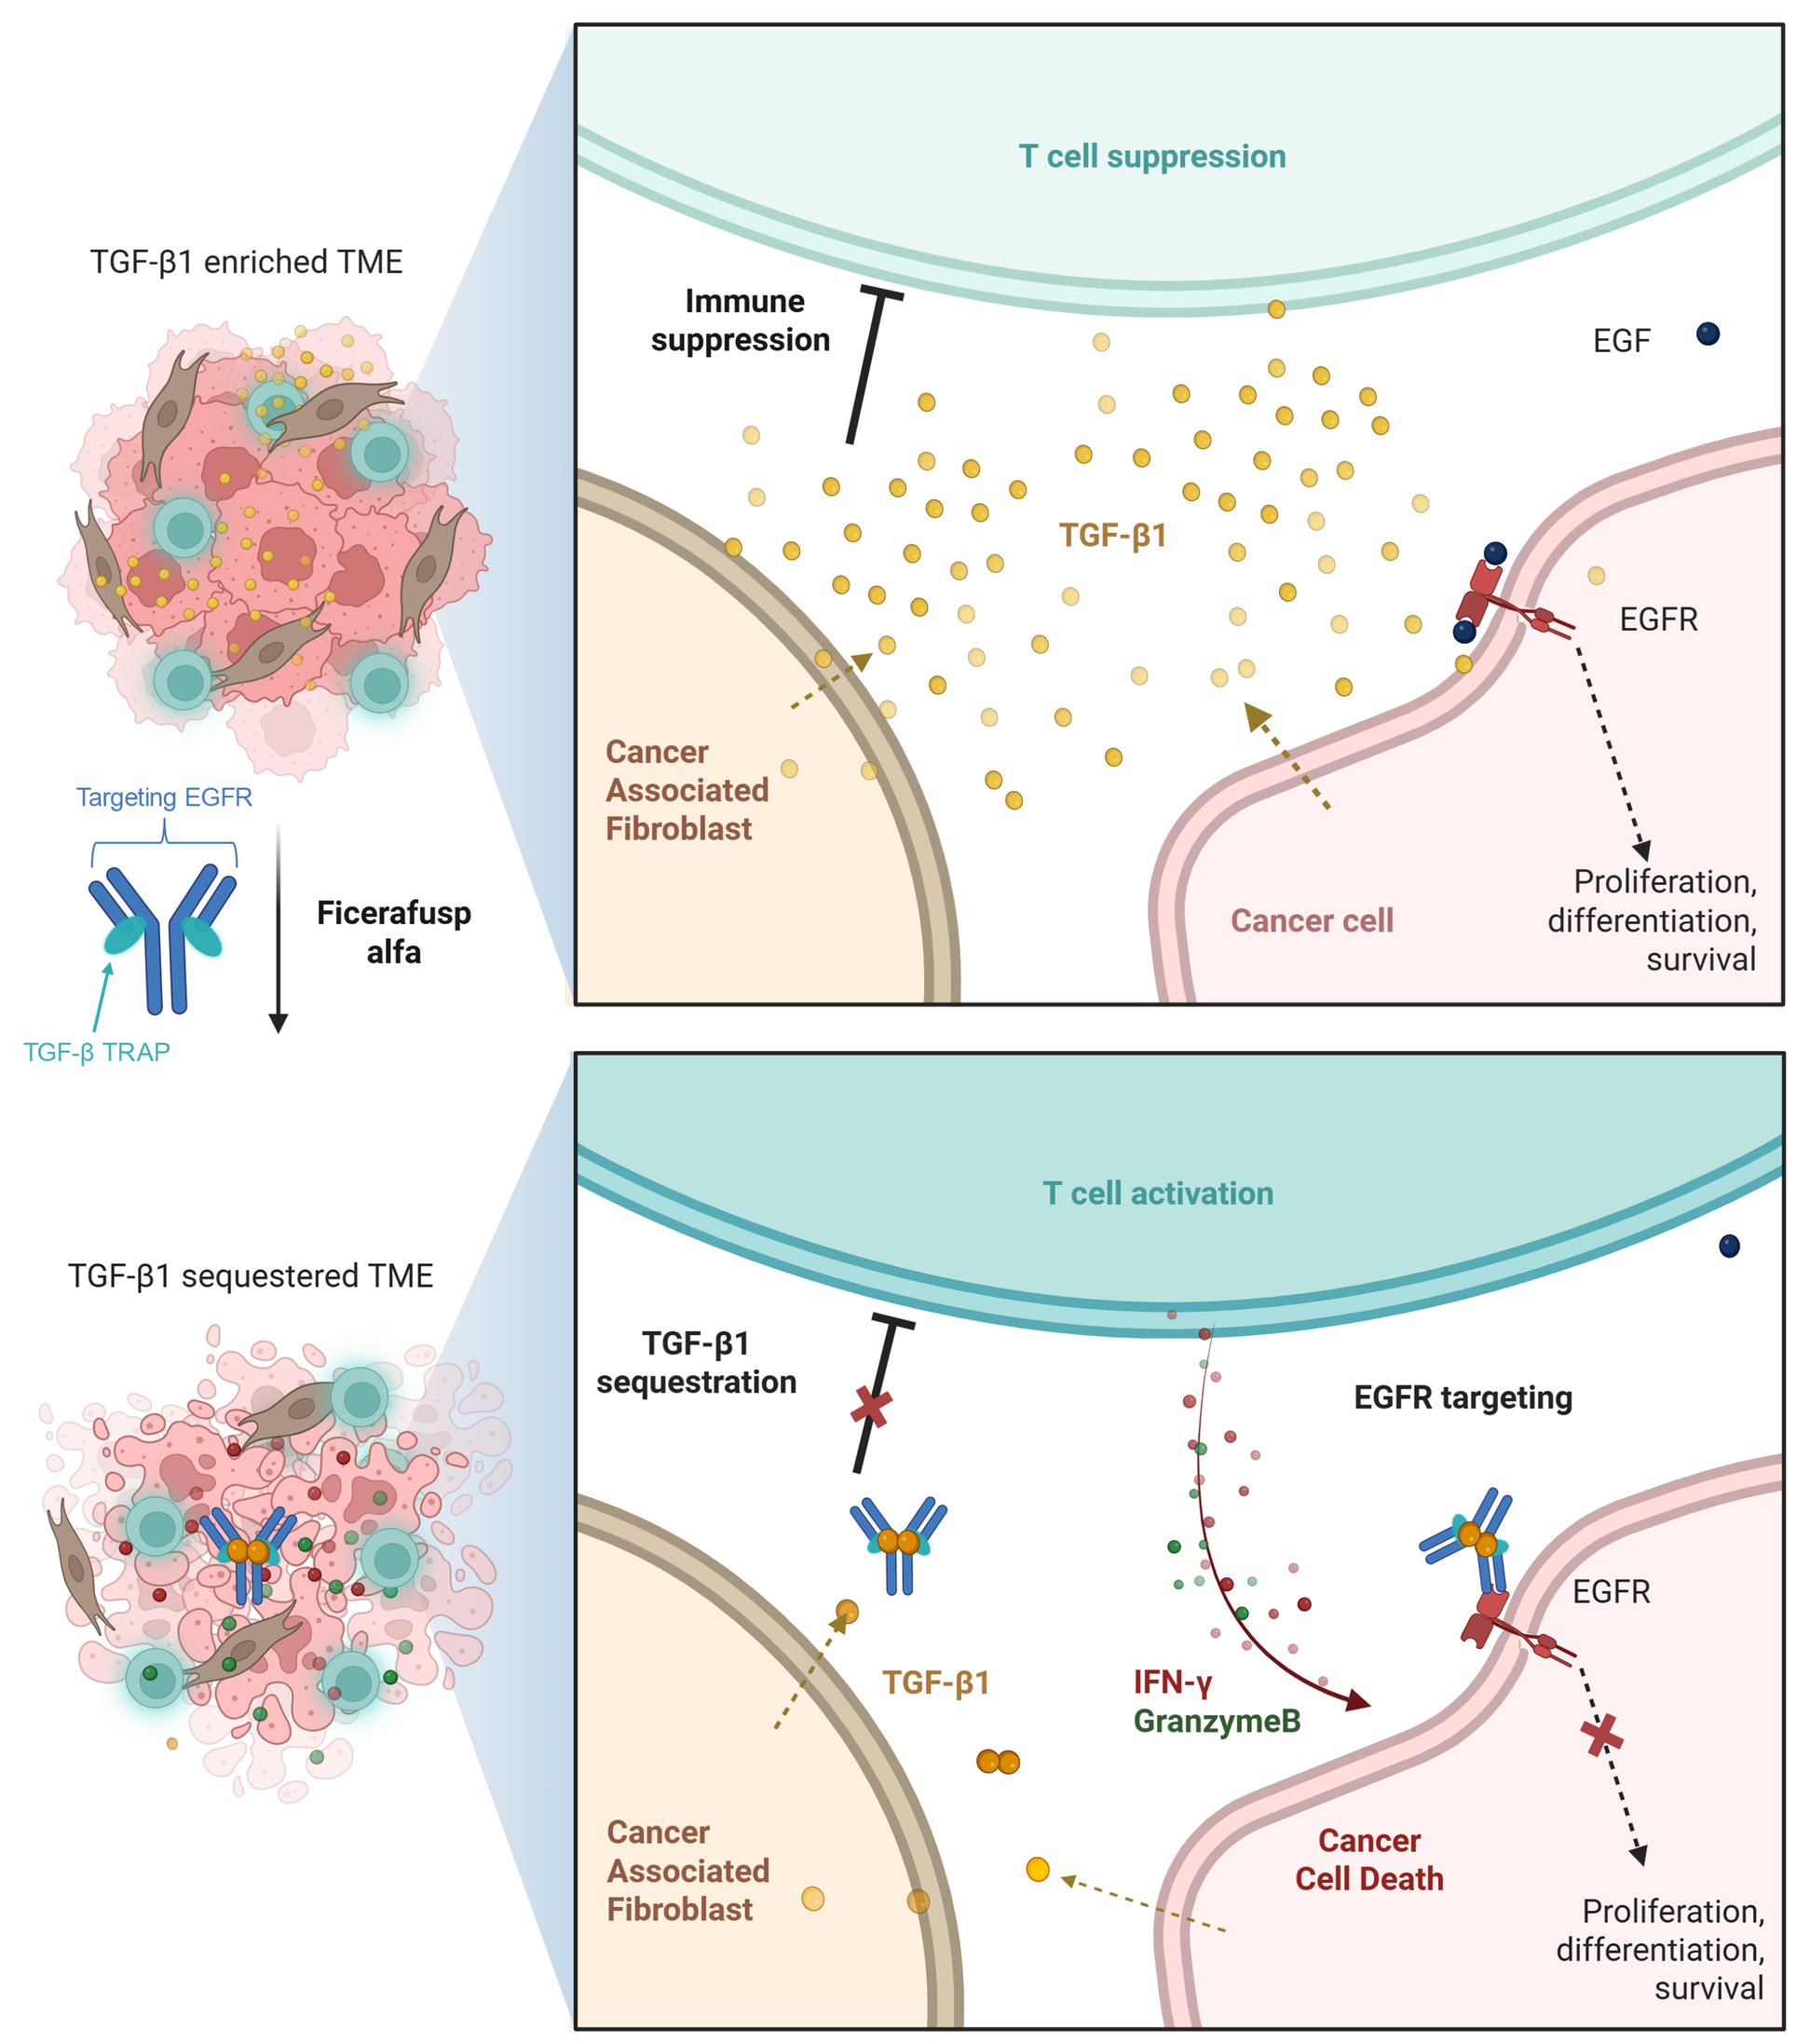
TGF-β1 enriched TME
T cell suppression
Immune suppression
EGF
TGF-β1
EGFR
Cancer Associated Fibroblast
Cancer cell
Proliferation, differentiation, survival
Targeting EGFR
Ficerafusp alfa
TGF-β TRAP
TGF-β1 sequestered TME
T cell activation
TGF-β1 sequestration
EGFR targeting
TGF-β1
IFN-γ
GranzymeB
Cancer Cell Death

In anti-EGFR–sensitive CRC cell lines, ficerafusp alfa exerts antitumor activity by simultaneously blocking both cancer cell-intrinsic EGFR-mediated survival and proliferation, and immunosuppressive TGF-β1 signaling within the TME. In anti-EGFR–resistant CRC cell lines, even though blocking of downstream signaling is ineffective, the TME is targeted via the anti-EGFR arm, leading to effective and on-target TGF-β1 sequestration. Ficerafusp alfa rescues the inhibitory effect of TGF-β1, enhancing IFN-γ and granzyme B secretion and cytolytic activity of T cells against both EGFR-sensitive and -resistant CRC cell lines. This illustrates the therapeutic benefit of the TGF-β–targeting arm of ficerafusp alfa in overcoming immune suppression and anti-EGFR resistance in CRC. Figure created with BioRender.com.

BACKGROUND

- CRC is the third most prevalent cancer globally,[1] with ~20% of patients diagnosed with metastatic CRC (mCRC). Additionally, up to half of those initially diagnosed with localized tumors develop metastases,[2] among which ~95% are MSS tumors and 50% are *KRAS* WT
- First-line treatment involves chemotherapy plus anti-EGFR (left-sided CRC) or anti-VEGF (left- and right-sided CRC) agents[3,4] but efficacy of treatments is limited by tumor heterogeneity, acquired drug resistance, and a highly immunosuppressive TME associated with poor tumor penetration of immune cells[5-8]
- Four consensus molecular subtypes of CRC have been defined, with the CMS2 subtype associated with EGFR dependence and the CMS4 subtype associated with a TGF-β1–driven mesenchymal phenotype[9]
- Ficerafusp alfa, a bifunctional antibody targeting EGFR and TGF-β, was specifically designed to enable tumor penetration and overcome key resistance mechanisms seen with anti-PD-1 and anti-EGFR antibodies (eg, cetuximab) by blocking epithelial-mesenchymal transition (EMT) and reversing immune suppression[10,11]
- Ficerafusp alfa reduces VEGF and the angiogenesis pathway compared with cetuximab in mouse models and in combination with pembrolizumab in HNSCC paired tumor biopsies (NCT04429542)[10,11]
- In this study, ficerafusp alfa was evaluated in KRAS/BRAF WT and MSS CRC cell lines to determine if dual targeting of EGFR and TGF-β reverses the immunosuppressive effects of TGF-β and improves antitumor activity compared with cetuximab

RESULTS

Figure 2. In the absence of PBMCs, NCI-H508 cells are sensitive to ficerafusp alfa and cetuximab (anti-EGFR), whereas SNU-503 cells with high basal TGF-β1 expression are largely resistant.

A

Cell line	KRAS	BRAF-V600	Consensus molecular subtype	Microsatellite instability status
NCI-H508	WT	WT	CMS2	MSS
SNU-503	WT	WT	CMS4	MSS

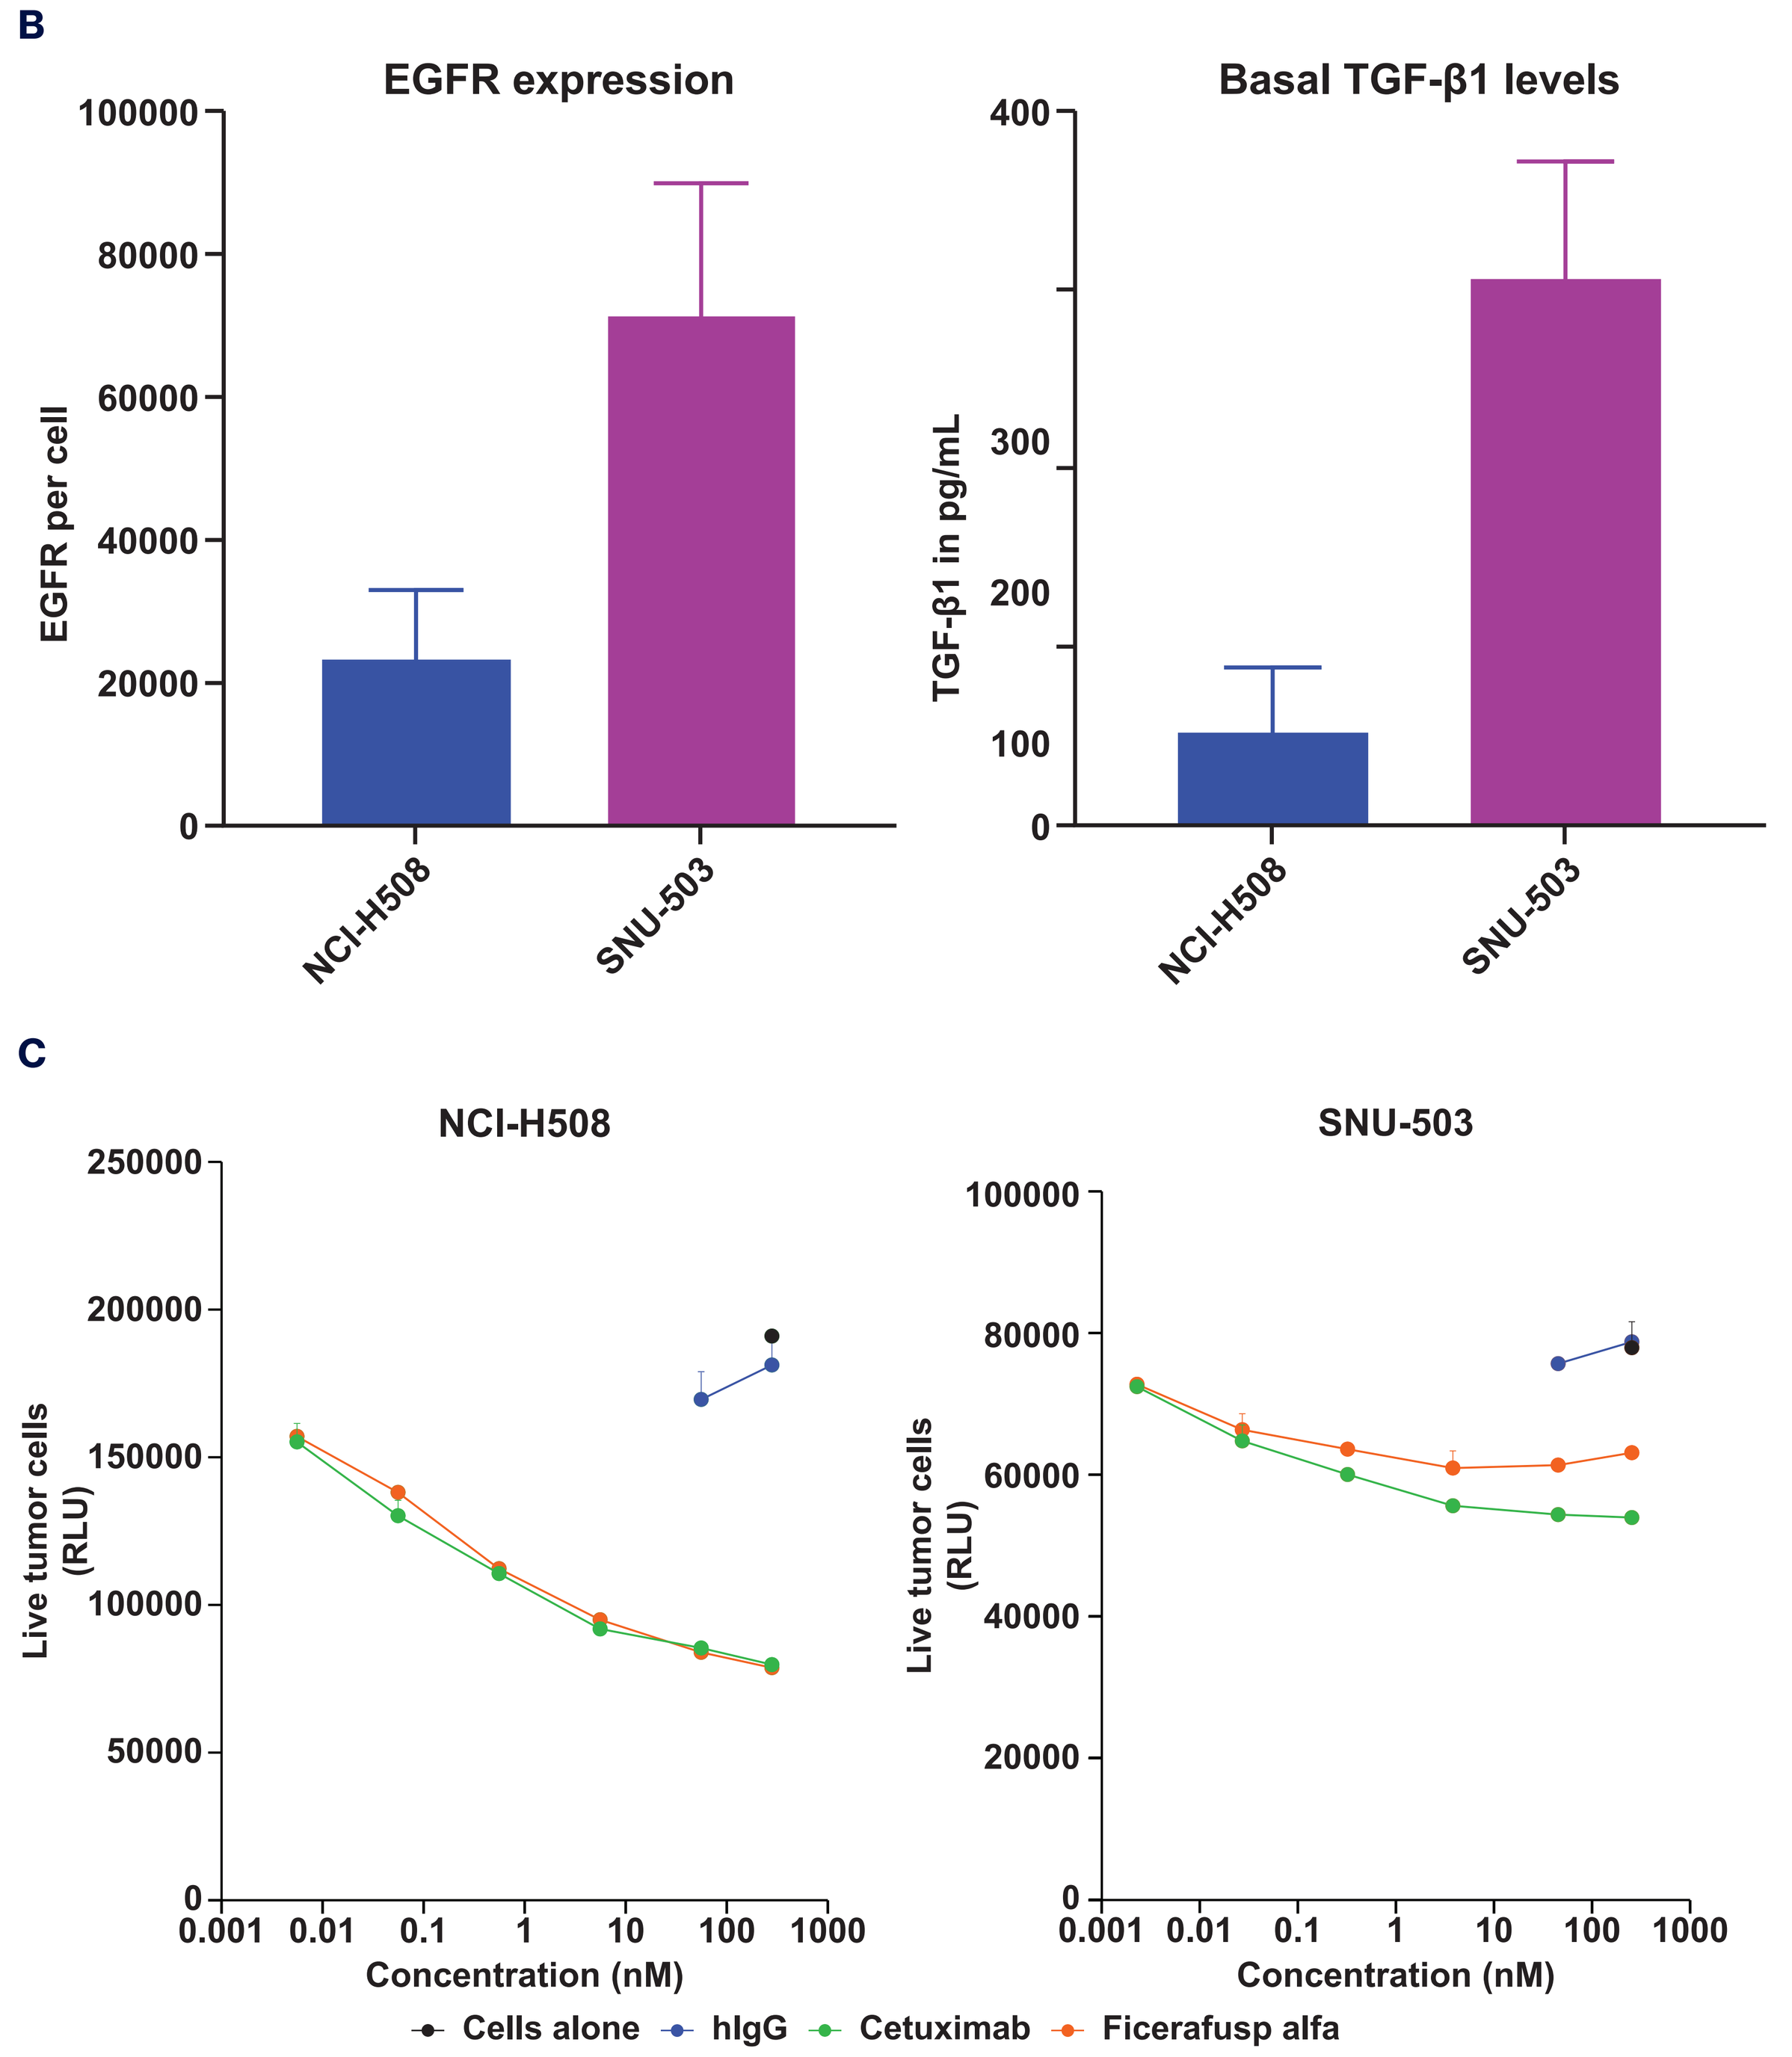
B
EGFR expression
EGFR per cell
NCI-H508
SNU-503
Basal TGF-β1 levels
TGF-β1 in pg/mL
C
NCI-H508
SNU-503
Live tumor cells (RLU)
Concentration (nM)
Cells alone
hIgG
Cetuximab
Ficerafusp alfa

(A) Table summarizing the characteristics of the NCI-H508 and SNU-503 CRC cell lines.[12-14] **(B)** EGFR expression was evaluated by flow cytometry (mean ± SD; n=2) and basal TGF-β1 secretion from 0.5 million cells after 72 h was evaluated using ELISA (mean ± SD; n=2). **(C)** In an inhibition of proliferation assay, different concentrations of ficerafusp alfa or cetuximab (or human IgG control) were added to NCI-H508 and SNU-503 cells in 96-well plates and incubated for 72 h. Cell viability was measured by CellTiter-Glo (Promega). Representative data shown from duplicate experiments.

Figure 3. In the presence of PBMCs, ficerafusp alfa rescued the inhibitory effect of TGF-β1, enhancing IFN-γ secretion and cytolytic activity of PBMCs against NCI-H508 and SNU-503 cells.

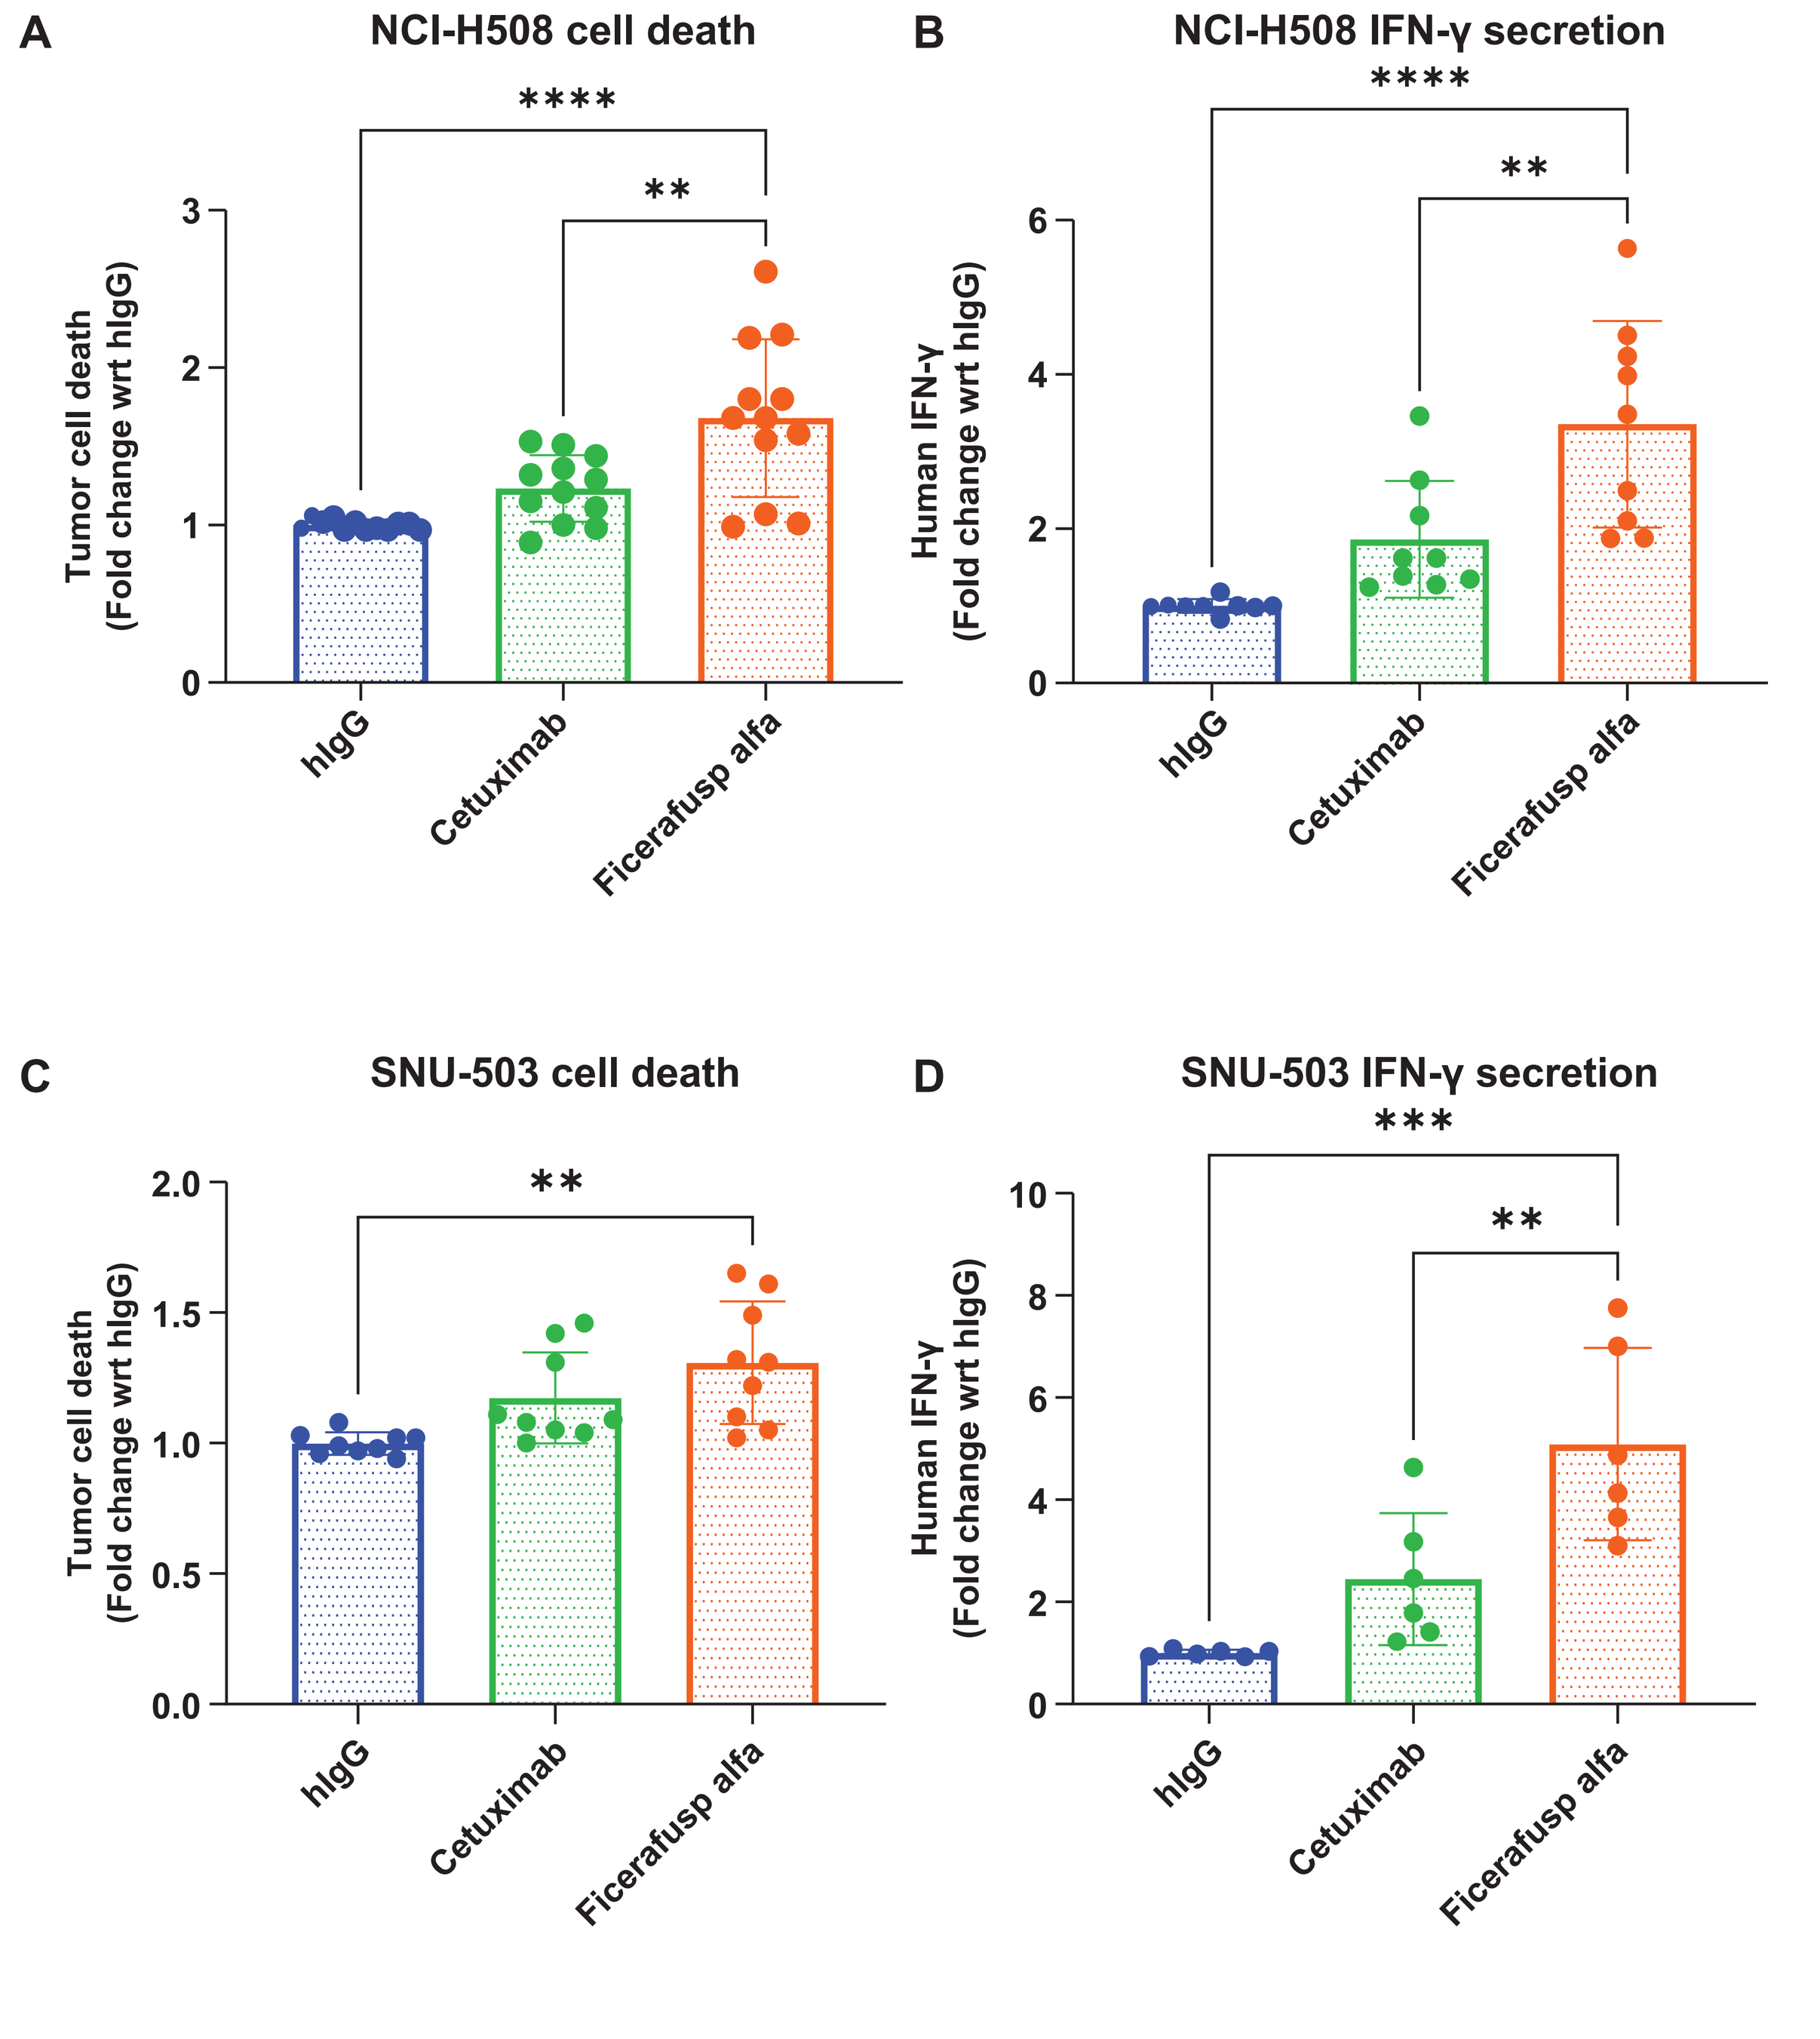
A
NCI-H508 cell death
Tumor cell death (Fold change wrt hIgG)
B
NCI-H508 IFN-γ secretion
Human IFN-γ (Fold change wrt hIgG)
C
SNU-503 cell death
D
SNU-503 IFN-γ secretion
hIgG
Cetuximab
Ficerafusp alfa

In tumor cell-PBMC coculture assays, human PBMCs prestimulated with soluble anti-CD3 antibody (2 ng/mL) and anti-CD28 antibody (10 ng/mL) were cocultured with NCI-H508 cells **(A and B)** and SNU-503 cells **(C and D)** in the presence of TGF-β1 (1 ng/mL). Human IgG, cetuximab, and ficerafusp alfa were added to a final concentration of 56 nM. After 48-72 h, CytoToxGlo readout was taken to measure dead cell proteases **(A and C)** and supernatant was evaluated for IFN-γ levels using ELISA **(B and D)**. The ratio of tumor cells:PMBCs was 1:10, and at least 2-3 different PBMC donors were evaluated. Data (mean ± SD) are plotted as fold change compared with the human IgG control group. Statistical analysis was done using a parametric one-way ANOVA multiple comparison test comparing ficerafusp alfa with other groups. Significance level: *P≤0.05; **P≤0.01; ***P≤0.001; ****P≤0.0001.

Figure 4. Ficerafusp alfa showed enhanced and sustained tumor cell cytotoxicity compared with cetuximab.

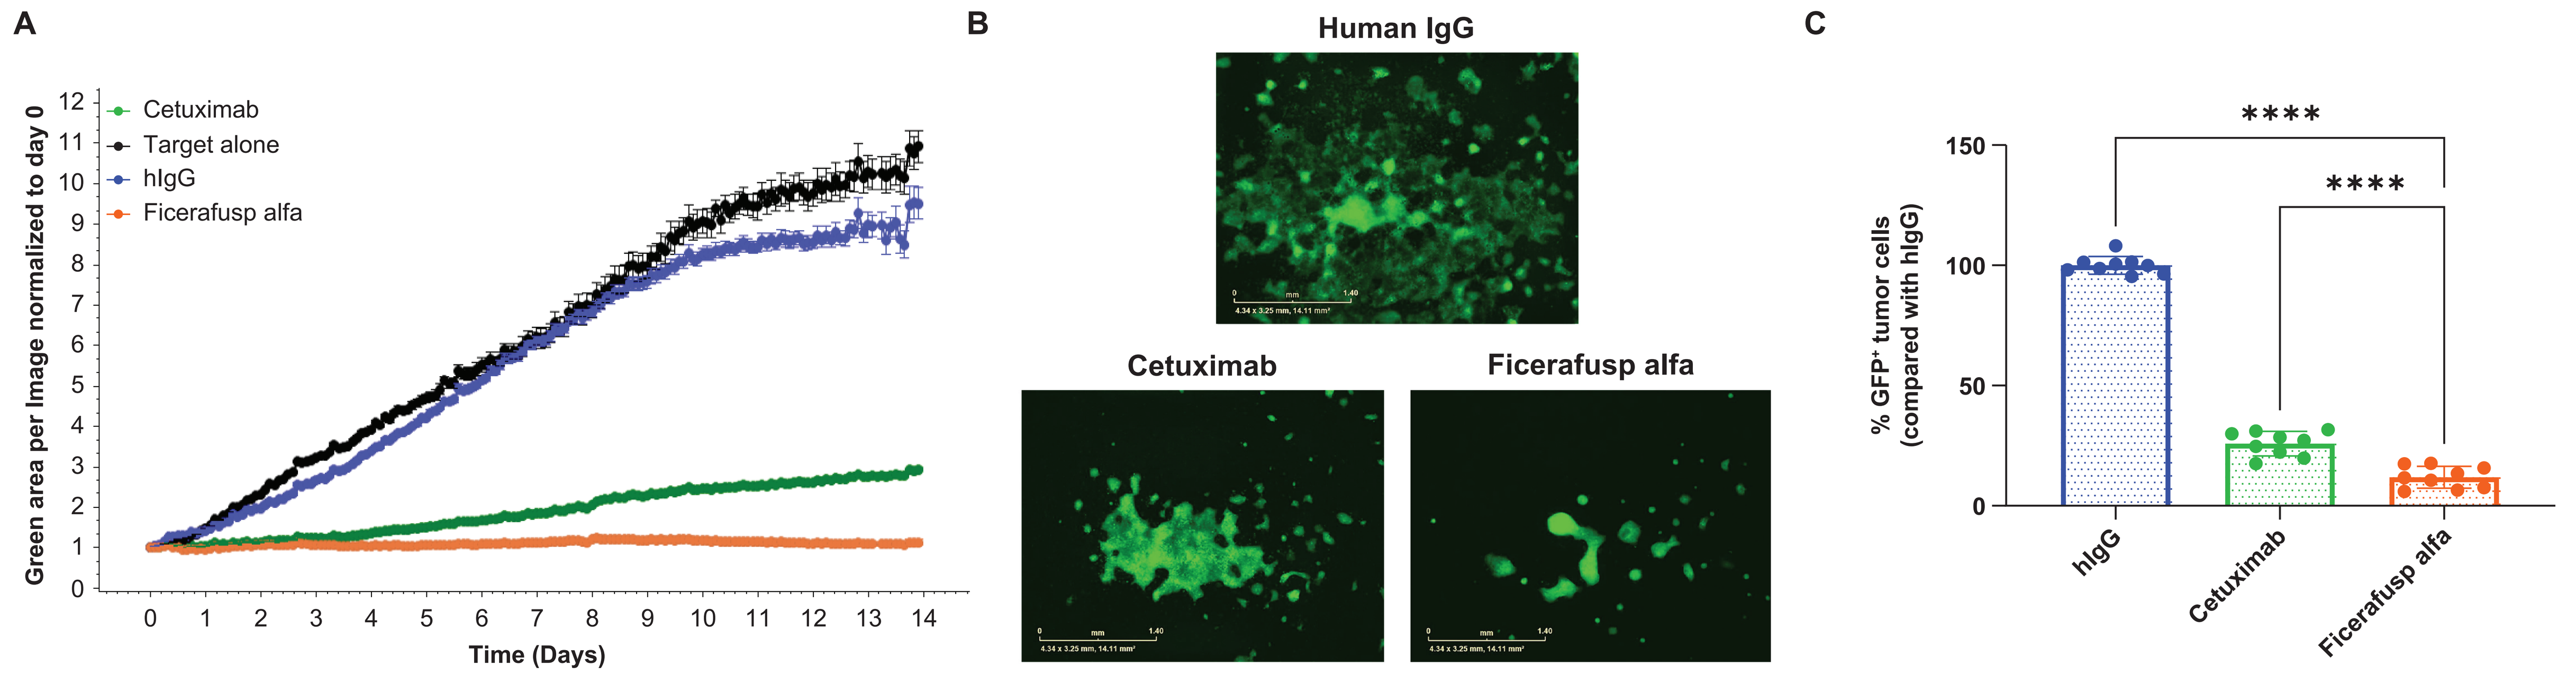
A
Cetuximab
Target alone
hIgG
Ficerafusp alfa
Green area per Image normalized to day 0
Time (Days)
B
Human IgG
Cetuximab
Ficerafusp alfa
C
% GFP+ tumor cells (compared with hIgG)
hIgG

Human PBMCs prestimulated with soluble anti-CD3 antibody (2 ng/mL) and anti-CD28 antibody (10 ng/mL) were cocultured with SNU-503 rfLuc-GFP cells in the presence of TGF-β (1 ng/mL). Human IgG, cetuximab, and ficerafusp alfa were added to a final concentration of 56 nM. Real-time images of GFP+ SNU-503 cells were captured at 4× magnification using the IncuCyte platform for 14 days. **(A)** Data plotted as green area per image normalized to day 0 (mean ± SEM). **(B)** Representative images of GFP+ SNU-503 cells on day 14. **(C)** Percentage of GFP+ SNU-503 cells compared with the human IgG control group on day 14. Data plotted from 3 independent experiments with 3 different PBMC donors (mean ± SD). Statistical analysis was done using a parametric one-way ANOVA multiple comparison test comparing ficerafusp alfa with other groups. Significance level: *P≤0.05; **P≤0.01; ***P≤0.001; ****P≤0.0001.

Figure 5. In the presence of CAF-conditioned media (CAF-CM), ficerafusp alfa showed significantly improved tumor cytotoxicity, enhanced expression of proinflammatory cytokines/chemokines, and reduced expression of protumor factors compared with cetuximab.

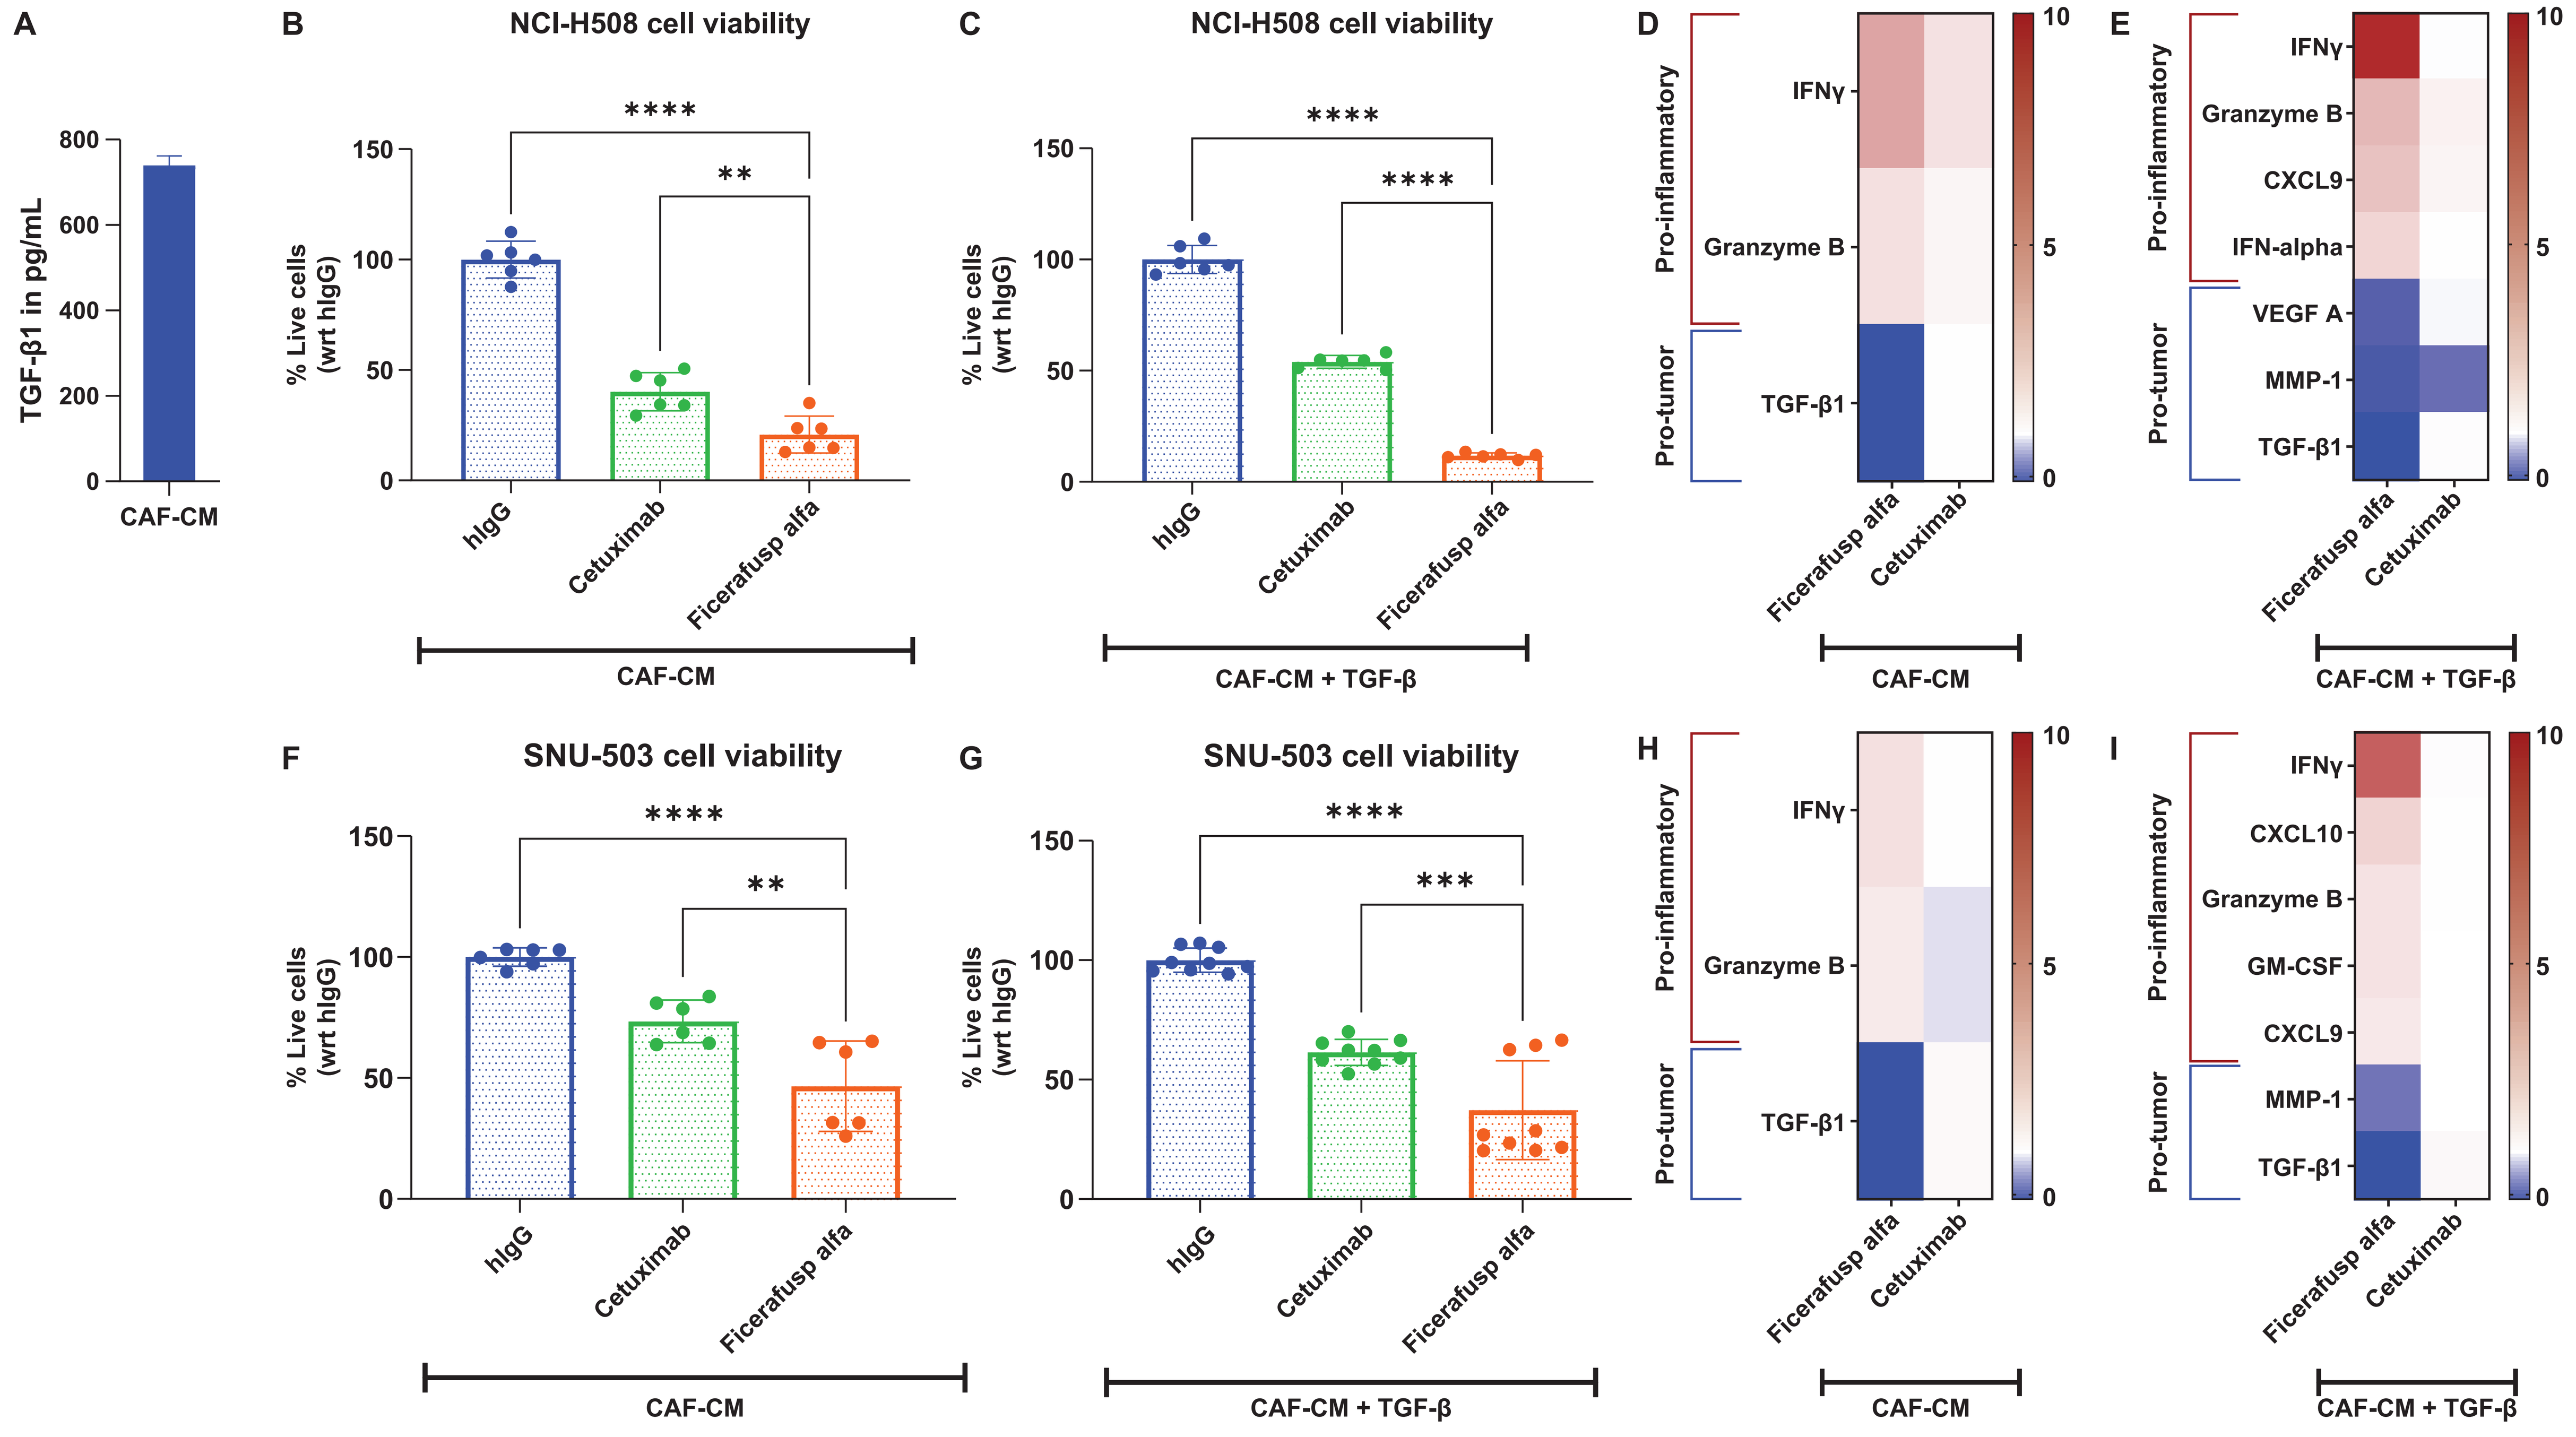
A
TGF-β1 in pg/mL
CAF-CM
B
NCI-H508 cell viability
% Live cells (wrt hIgG)
C
NCI-H508 cell viability
CAF-CM + TGF-β
D
Pro-inflammatory
Pro-tumor
IFNγ
Granzyme B
TGF-β1
E
CXCL9
IFN-alpha
VEGF A
MMP-1
F
SNU-503 cell viability
G
SNU-503 cell viability
H
I
CXCL10
GM-CSF
hIgG
Cetuximab
Ficerafusp alfa

(A) TGF-β1 secretion (mean ± SD) in FAP-positive CRC CAFs. 0.5 million CAFs were seeded and supernatant was collected after 72 h. TGF-β1 secretion was evaluated using ELISA and data were normalized to CAF growth media control. **(B-I)** In coculture assays, human PBMCs prestimulated with soluble anti-CD3 antibody (2 ng/mL) and anti-CD28 antibody (10 ng/mL) were cocultured with NCI-H508 **(B-E)** and SNU-503 **(F-I)** rfLuc-GFP cells in the presence of 50% CAF-CM ± TGF-β1 (1 ng/mL). Human IgG, cetuximab, and ficerafusp alfa were added at a final concentration of 56 nM. After incubation for 6 days, BioGlo readout was taken to measure luciferase-positive cells (tumor cells), and cytokines/chemokines in the supernatant were measured using the Human Premixed Multi-Analyte kit (Luminex assay; R&D Systems) and ELISA. The ratio of tumor cells:PMBCs was 1:10, and at least 2-3 different PBMC donors were evaluated. **(B, C, F, and G)** show percentage of live tumor cells (mean ± SD) compared with the human IgG control group. Statistical analysis was done using a parametric one-way ANOVA multiple comparison test comparing ficerafusp alfa with other groups. Significance level: *P≤0.05; **P≤0.01; ***P≤0.001; ****P≤ 0.0001. **(D, E, H, and I)** show Luminex/ELISA data. Values (pg/mL) were changed to fold change compared with the human IgG group. Analytes that showed ≥1.75× or <0.5× change compared with human IgG were plotted as a heat map. Statistical analysis was done using an unpaired t test, and analytes with a statistically significant difference (P≤0.05) for cetuximab compared with ficerafusp alfa are shown.

REFERENCES

1. Siegel RL, et al. *CA Cancer J Clin.* 2025;75(1):10-45.
2. Ciardiello F, et al. *CA Cancer J Clin.* 2022;72(4):372-401.
3. Gmeiner WH. *Cancers (Basel).* 2024;16(5).
4. Cherri S, et al. *Front Oncol.* 2023;13:1246716.
5. Parseghian C, et al. *Front Cell Dev Biol.* 2023;11:1176657.
6. Schmitt M and Greten FR. Nat Rev Immunol. 2021;21(10):653-67.
7. Dagogo-Jack I and Shaw AT. Nat Rev Clin Oncol. 2018;15(2):81-94.
8. Buikhuisen JY, et al. *Oncogenesis.* 2020;9(7):66.
9. Martini G, et al. *Ther Adv Med Oncol.* 2020;12:1758835920936089.
10. Boreddy SR, et al. *Cancer Res.* 2023;83(11):1883-904.
11. O'Connell BC, et al. *Ann Oncol.* 2025;36(Suppl 2):abstract 269eP.
12. Berg KCG, et al. *Mol Cancer.* 2017;16(1):116.
13. Medico E, et al. *Nat Commun.* 2015;6:7002.
14. Ronen J, et al. *bioRxiv.* 2018;doi.org/10.1101/464743.

DISCLOSURES

BC O'Connell and **RL Salazar** are employees of Bicara Therapeutics Inc. **A Kuriakose**, **R Nair**, and **P Nair**, are employees of Syngene International Ltd, which was contracted by Bicara Therapeutics for the work reported.

ACKNOWLEDGMENTS

This study was funded by Bicara Therapeutics Inc. Medical writing support was provided by Nucleus Global, funded by Bicara Therapeutic Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SITC or the author of this poster.

SITC Annual Meeting | National Harbor, MD, USA | November 5-9, 2025

Contact: Brenda C O'Connell; email: brenda.o'connell@bicara.com